Durable responses with intratumoral electroporation of plasmid interleukin-12 plus pembrolizumab in patients with advanced triple-negative breast cancer: Cohort 1 update from KEYNOTE-890

Melinda L. Telli[1], Irene Wapnir[1], Bianca Devitt[2], Katharine Cuff[3], Hatem Soliman[4], Shaveta Vinayak[5], David A. Canton[6], Christopher Twitty[6], Sunny Xie[6], Ying Lu[6], Donna Bannavong[6], Bridget O'Keeffe[6], Sandra Aung[6], Rohit Joshi[7]

[1]Stanford University School of Medicine, Stanford, CA, USA; [2]Box Hill Hospital, Box Hill, VIC, Australia; [3]Princess Alexandra Hospital, Woolloongabba, QLD, Australia; [4]Moffit Cancer Center, Tampa, FL, USA; [5]University of Washington, Fred Hutchinson Cancer Research Center, Seattle Cancer Care Alliance, Seattle, WA, USA; [6]OncoSec Medical Incorporated, San Diego, CA, USA; [7]Adelaide Oncology and Haematology, Adelaide, SA, Australia

San Antonio Breast Cancer Symposium® December 7-10, 2021

P2-14-06

Background

- Plasmid interleukin-12 (tavokinogene telseplasmid; TAVO) delivered to accessible tumors by intratumoral injection and electroporation (EP) induces sustained local expression of IL-12 (intratumor levels of IL-12 higher than baseline for up to weeks).[1]
- IL-12 is a potent immunoregulatory cytokine that plays a key role in the crosstalk between innate (dendritic, macrophage, and natural killer) and adaptive (T and B) cells, promoting anti-tumor immune responses.[2]
- TAVO-EP has been shown to induce activation of innate and adaptive tumor-infiltrating and peripheral immune cells, regression of treated and distant untreated lesions (abscopal effect), and expression of PD-L1 in patients with melanoma and TNBC, without the systemic toxicity that limited therapeutic use of IL-12 historically.[3]
- The combination of TAVO-EP and pembrolizumab has demonstrated durable responses in melanoma patients with immunologically "cold" tumors or with prior progression on anti-PD1 therapy.[4]
- Anti-PD1 monotherapy has 5-10% overall response rate (ORR) in the second-line or later (2L+) treatment setting for advanced TNBC.[5]
- New antibody-directed conjugate (ADC) chemotherapy has increased rates of responses compared with prior standard chemotherapy in 2L+ advanced TNBC; however, short duration of response (DOR) and toxicity are issues of concern.[6]
- Therapies that can induce durable responses with limited toxicity are needed.

Mechanism of Action of TAVO-EP

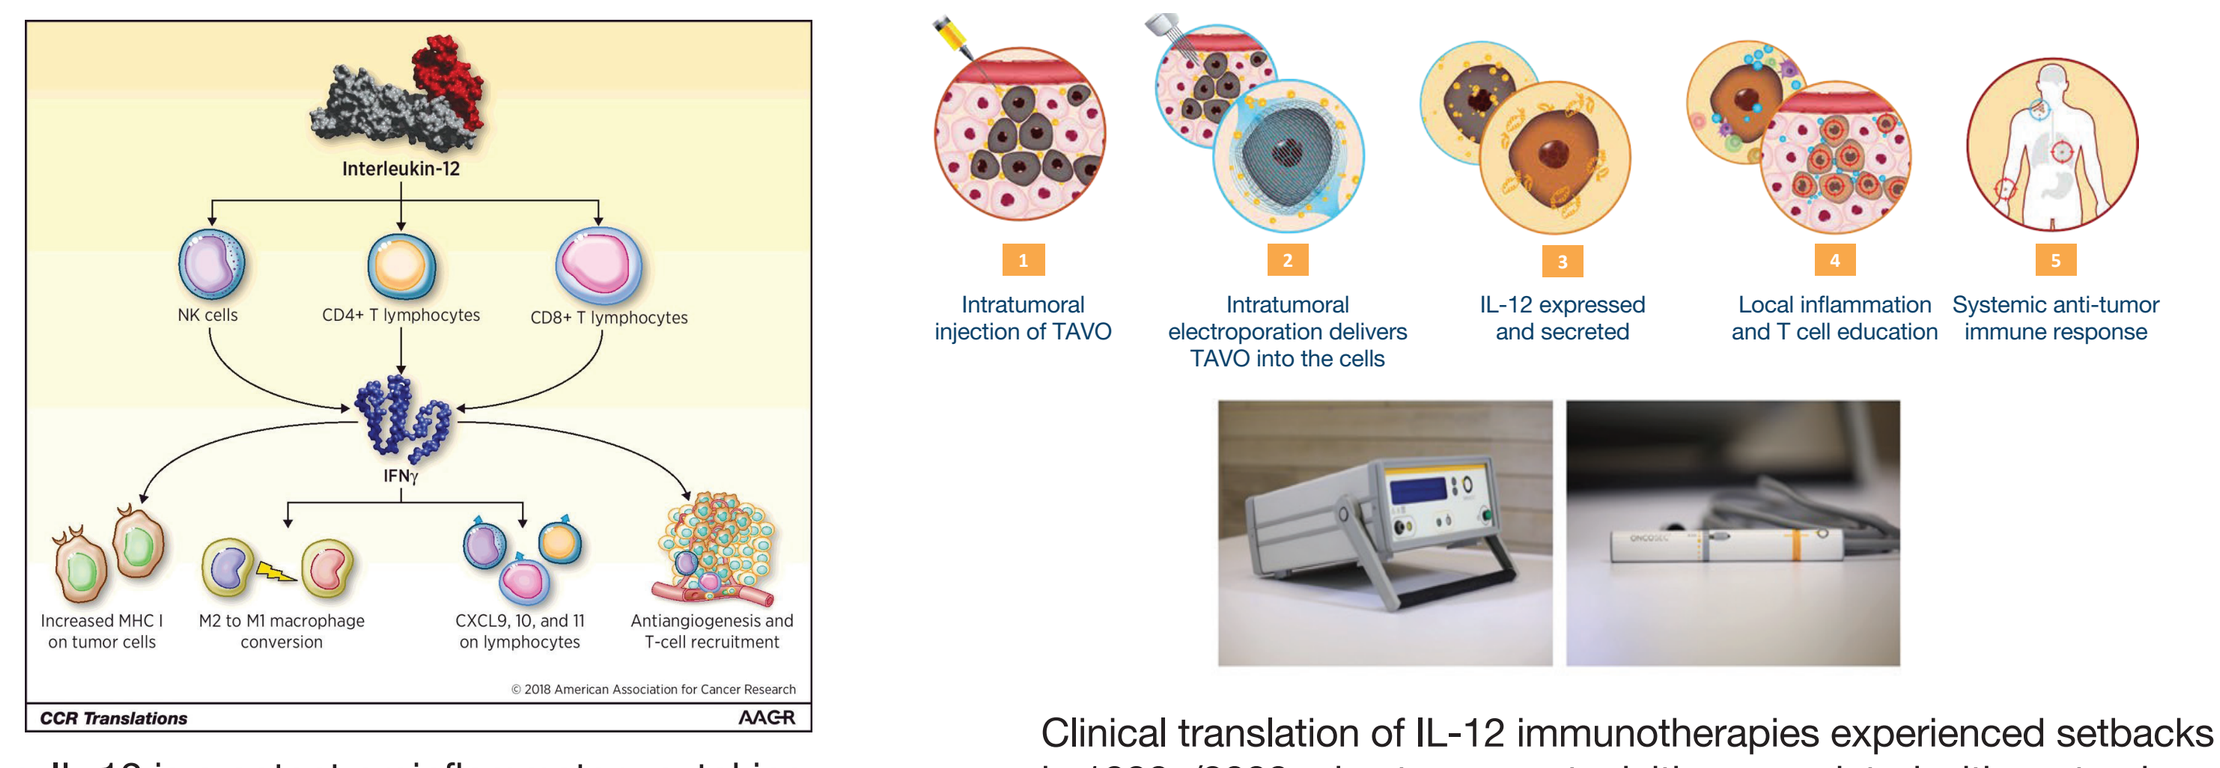
Interleukin-12
NK cells
CD4+ T lymphocytes
CD8+ T lymphocytes
IFNγ
Increased MHC I on tumor cells
M2 to M1 macrophage conversion
CXCL9, 10, and 11 on lymphocytes
Antiangiogenesis and T-cell recruitment
© 2018 American Association for Cancer Research
CCR Translations
AACR
1 Intratumoral injection of TAVO
2 Intratumoral electroporation delivers TAVO into the cells
3 IL-12 expressed and secreted
4 Local inflammation and T cell education
5 Systemic anti-tumor immune response

- IL-12 is a potent proinflammatory cytokine with the following antitumor mechanisms:
 - Increasing MHC I antigen presentation
 - Attracting additional NK, Th1, and CD8+ T cells into TME
 - Reversing tumor-induced immunosuppression

Clinical translation of IL-12 immunotherapies experienced setbacks in 1990s/2000s due to severe toxicities associated with systemic IL-12 injections. Intratumoral delivery of plasmid IL-12 (TAVO) followed by electroporation yields sustained expression of IL-12 and stimulates a systemic proinflammatory immune response, without systemic immune-related toxicities.

Methods

- Cohort 1 of this Phase 2, open-label, multicenter study assessed the safety and efficacy of TAVO-EP in combination with pembrolizumab as 2L+ treatment for advanced TNBC.
- Eligible patients had at least 1 line of prior systemic therapy for advanced or metastatic disease, measurable disease by RECIST v1.1, and ≥1 lesion accessible for TAVO-EP treatment.
- Patients received pembrolizumab (200 mg IV) every 3 weeks and TAVO-EP (0.5 mg/mL at dose volume of 1/4 lesion volume) on Days 1, 5, and 8 every 6 weeks. Patients could be treated beyond RECIST v.1.1 progression if patient was clinically stable until confirmed progression by iRECIST (iCPD).
- Tumor assessments were performed every 12 weeks.
- The primary endpoint was RECIST v.1.1 ORR by investigator review.
- Secondary endpoints included safety and tolerability, DOR, progression-free survival, immune-related RECIST (iRECIST) ORR and PFS, disease control rate, and overall survival (OS).

ClinicalTrials.gov: NCT03567720.

KEYNOTE-890: Study Design and Conduct[7]

Patients

COHORT 1 (N=26)
Patients with previously treated inoperable locally advanced or metastatic TNBC

COHORT 2 (N=40)[a]
Treatment-naïve patients with inoperable locally advanced or metastatic TNBC

[a]Recruiting
[b]Based on RECIST v1.1 unless specified
[c]Blinded Independent Central Review

Treatment Schedule

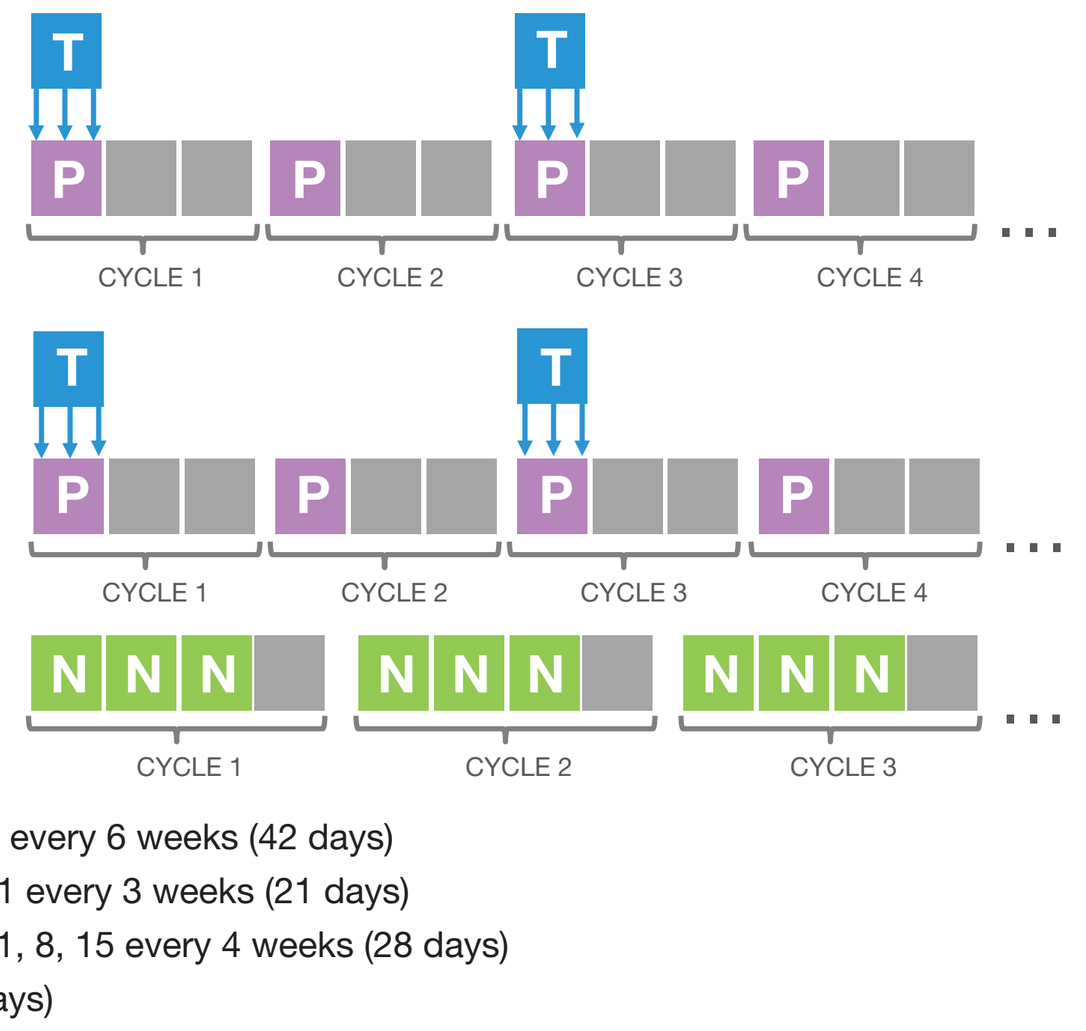
T
P
CYCLE 1
CYCLE 2
CYCLE 3
CYCLE 4
N
CYCLE 1
CYCLE 2
CYCLE 3

- T Tavo-EP on Days 1, 5, 8 every 6 weeks (42 days)
- P Pembrolizumab on Day 1 every 3 weeks (21 days)
- N Nab-paclitaxel on Days 1, 8, 15 every 4 weeks (28 days)
- Represents 1 week (7 days)

Key Endpoints

PRIMARY[b]
- COHORT 1: ORR by Investigator
- COHORT 2: ORR by BICR[c]

SECONDARY
- COHORT 1 (by Investigator)[b]: DOR, PFS, DCR and iPFS, iORR based on iRECIST
- COHORT 2: DOR, PFS, DCR and iPFS, iORR based on iRECIST by BICR and ORR by Investigator
- OS for all patients

EXPLORATORY ENDPOINTS
- Immune monitoring correlates with responders and non-responders
- Estimate of preliminary ORR in PD-L1 negative patients

Results

Patient Demographics and Disease Characteristics

Characteristic		N=26
Age	Median (range)	51.5 (30, 70)
Race	White	19 (73.1)
	Black or African American	3 (11.5)
	Asian	1 (3.8)
	Native Hawaiian or Other Pacific Islander	1 (3.8)
	Other	2 (7.7)
ECOG PS	0	16 (61.5)
	1	10 (38.5)
PD-L1 status	Negative (CPS <1)	3 (11.5)
	Low (CPS 1 to 9)	8 (30.8)
	High (CPS ≥10)	4 (15.4)
	Unknown	11 (42.3)
Prior lines of metastatic therapy	0	0 (0.0)
	1	9 (34.6)
	2	7 (26.9)
	3+	
Prior systemic therapy	taxane	22 (84.6)
	anthracycline	21 (80.8)
	cyclophosphamide	21 (80.8)
	capecitabine	15 (57.7)
	platinum therapy	14 (53.8)
	eribulin	8 (30.8)
	hormone therapy	8 (30.8)
	gemcitabine	6 (23.1)
	anti-PD(L)-1	4 (15.4)
	PARP inhibitor	4 (15.4)
Prior radiation therapy	Yes	19 (73.1)
Sites of disease	Lymph node	24 (92.3)
	Lung or liver	14 (53.8)
	- Lung	11 (42.3)
	- Liver	6 (23.1)
	Breast	12 (46.2)
	Skin	10 (38.5)
	Chest wall	10 (38.5)
	Bone	7 (26.9)
Median number of lesions (range)		5.5 (1-16)

Disposition

Between 01Nov2018 and 30Jan2020, 26 patients were enrolled and received at least one dose of study treatment (median follow up of 11.1 months).

Patients enrolled N=26
Completed treatment N=2
Discontinued treatment N=24

Disease progression	Adverse event	Patient decision	Investigator decision	Withdrawn consent	Other
Pembro: 14 (54%) TAVO EP: 13 (50%)	Pembro: 5 (19%) TAVO EP: 4 (15%)	Pembro: 1 (4%) TAVO EP: 1 (4%)	Pembro: 3 (12%) TAVO EP: 3 (12%)	Pembro: 1 (4%) TAVO EP: 1 (4%)	Pembro: 2 (8%) TAVO EP: 4 (15%)

Treatment-related Adverse Events

	Any Grade N (%)	Grade 1-2 N (%)	Grade 3* N (%)
Any event	26 (100)	20 (76.9)	6 (23.1)
Preferred Term			
Administration site pain	17 (65.4)	17 (65.4)	–
Fatigue	8 (30.8)	5 (19.2)	3 (11.5)
Nausea	5 (19.2)	5 (19.2)	–
Hypothyroidism	4 (15.4)	4 (15.4)	–
Decreased appetite	3 (11.5)	3 (11.5)	–
Vomiting	3 (11.5)	3 (11.5)	–
Anxiety	2 (7.7)	2 (7.7)	–
Diarrhea	2 (7.7)	2 (7.7)	–
Dyspnea	2 (7.7)	2 (7.7)	–
Injection site pain	2 (7.7)	2 (7.7)	–
Injection site reaction	2 (7.7)	2 (7.7)	–
Weight decreased	2 (7.7)	2 (7.7)	–

*There were no Grade 4 or Grade 5 TRAEs, additional Grade 3 events included in 1 patient each acute kidney injury, enterocolitis, and myocarditis.

TEAEs in >5 Patients

	Any Grade N (%)	Grade 1-2 N (%)	Grade 3* N (%)
Any event	26 (100)	7 (26.9)	15 (57.7)
Preferred Term			
Administration site pain	17 (65.4)	17 (65.4)	–
Nausea	15 (57.7)	13 (50)	2 (7.7)
Fatigue	14 (53.8)	11 (42.3)	3 (11.5)
Dyspnea	10 (38.5)	9 (34.6)	1 (3.8)
Diarrhea	9 (34.6)	8 (30.8)	1 (3.8)
Vomiting	9 (34.6)	8 (30.8)	1 (3.8)
Cough	8 (30.8)	8 (30.8)	–
Constipation	7 (26.9)	6 (23.1)	1 (3.8)
Anxiety	6 (23.1)	6 (23.1)	–
Decreased appetite	6 (23.1)	6 (23.1)	–
Pain	6 (23.1)	3 (11.5)	3 (11.5)
Arthralgia	5 (19.2)	5 (19.2)	–
Headache	5 (19.2)	5 (19.2)	–
Hypothyroidism	5 (19.2)	5 (19.2)	–
Pruritis	5 (19.2)	5 (19.2)	–
Upper respiratory tract infection	5 (19.2)	5 (19.2)	–

*Reported Grade 5 events included disease progression in 3 patients; Grade 4 events included sepsis in 2 patients and hypoglycemia in 1 patient.

Best RECIST v1.1 Responses

Best Percent Reduction from Baseline

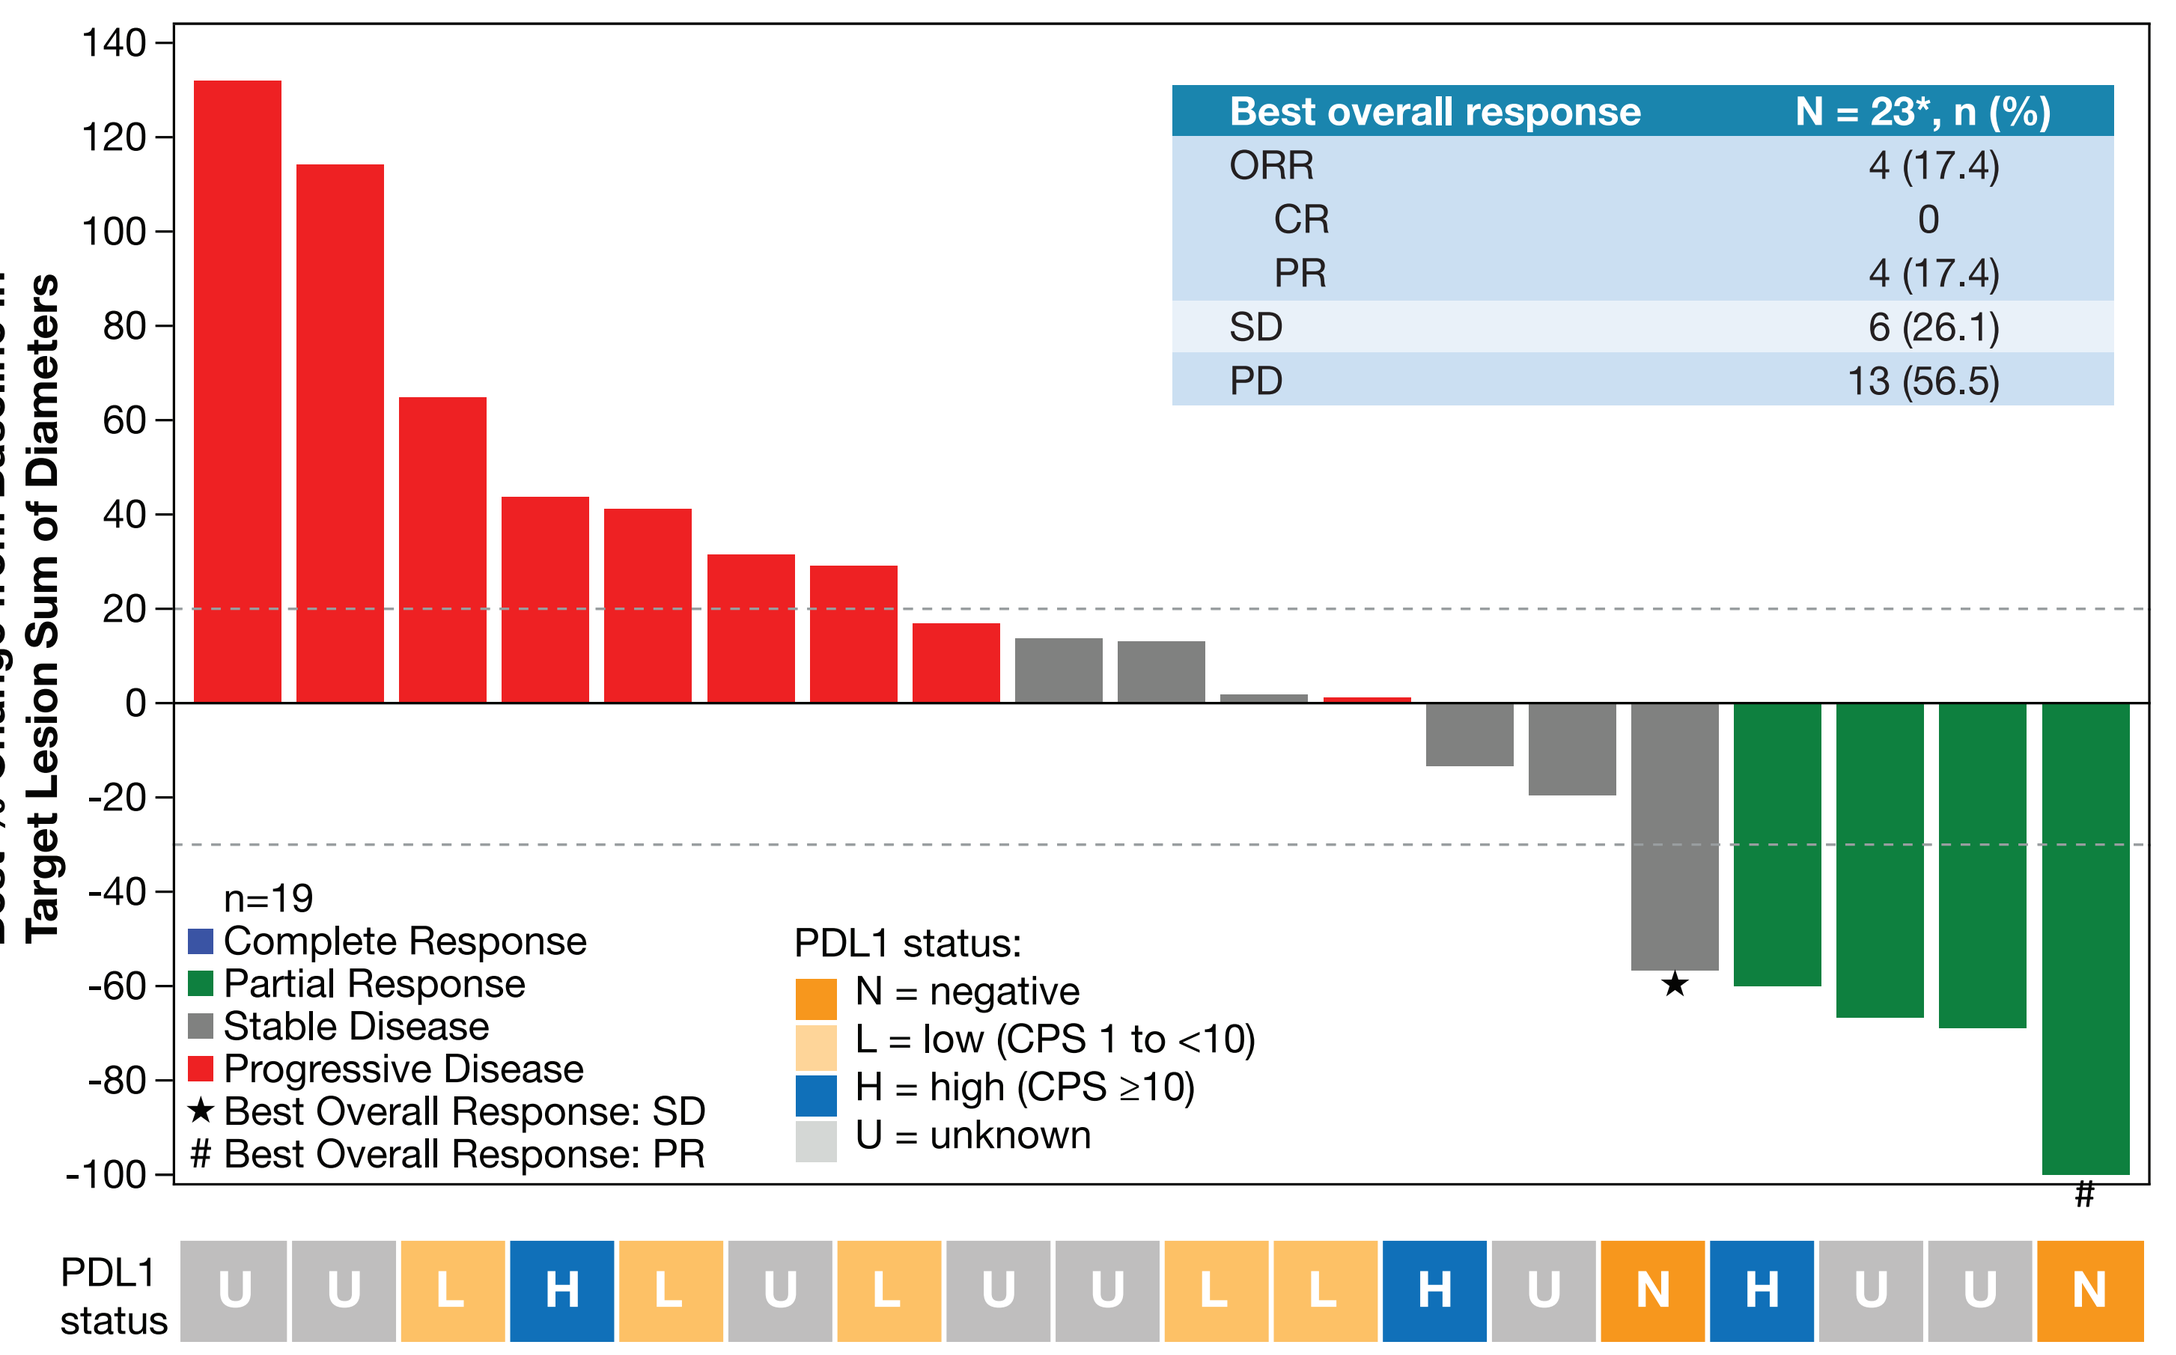
Best % Change from Baseline in Target Lesion Sum of Diameters
n=19
Complete Response
Partial Response
Stable Disease
Progressive Disease
★ Best Overall Response: SD
Best Overall Response: PR
PDL1 status:
N = negative
L = low (CPS 1 to <10)
H = high (CPS ≥10)
U = unknown
PDL1 status
U U L H L U L U U L L H U N H U U N

Best overall response	N = 23*, n (%)
ORR	4 (17.4)
CR	0
PR	4 (17.4)
SD	6 (26.1)
PD	13 (56.5)

Change in Target Lesion Sum of Lesion Diameters

Percent Change in Target Lesion SLD Over Time

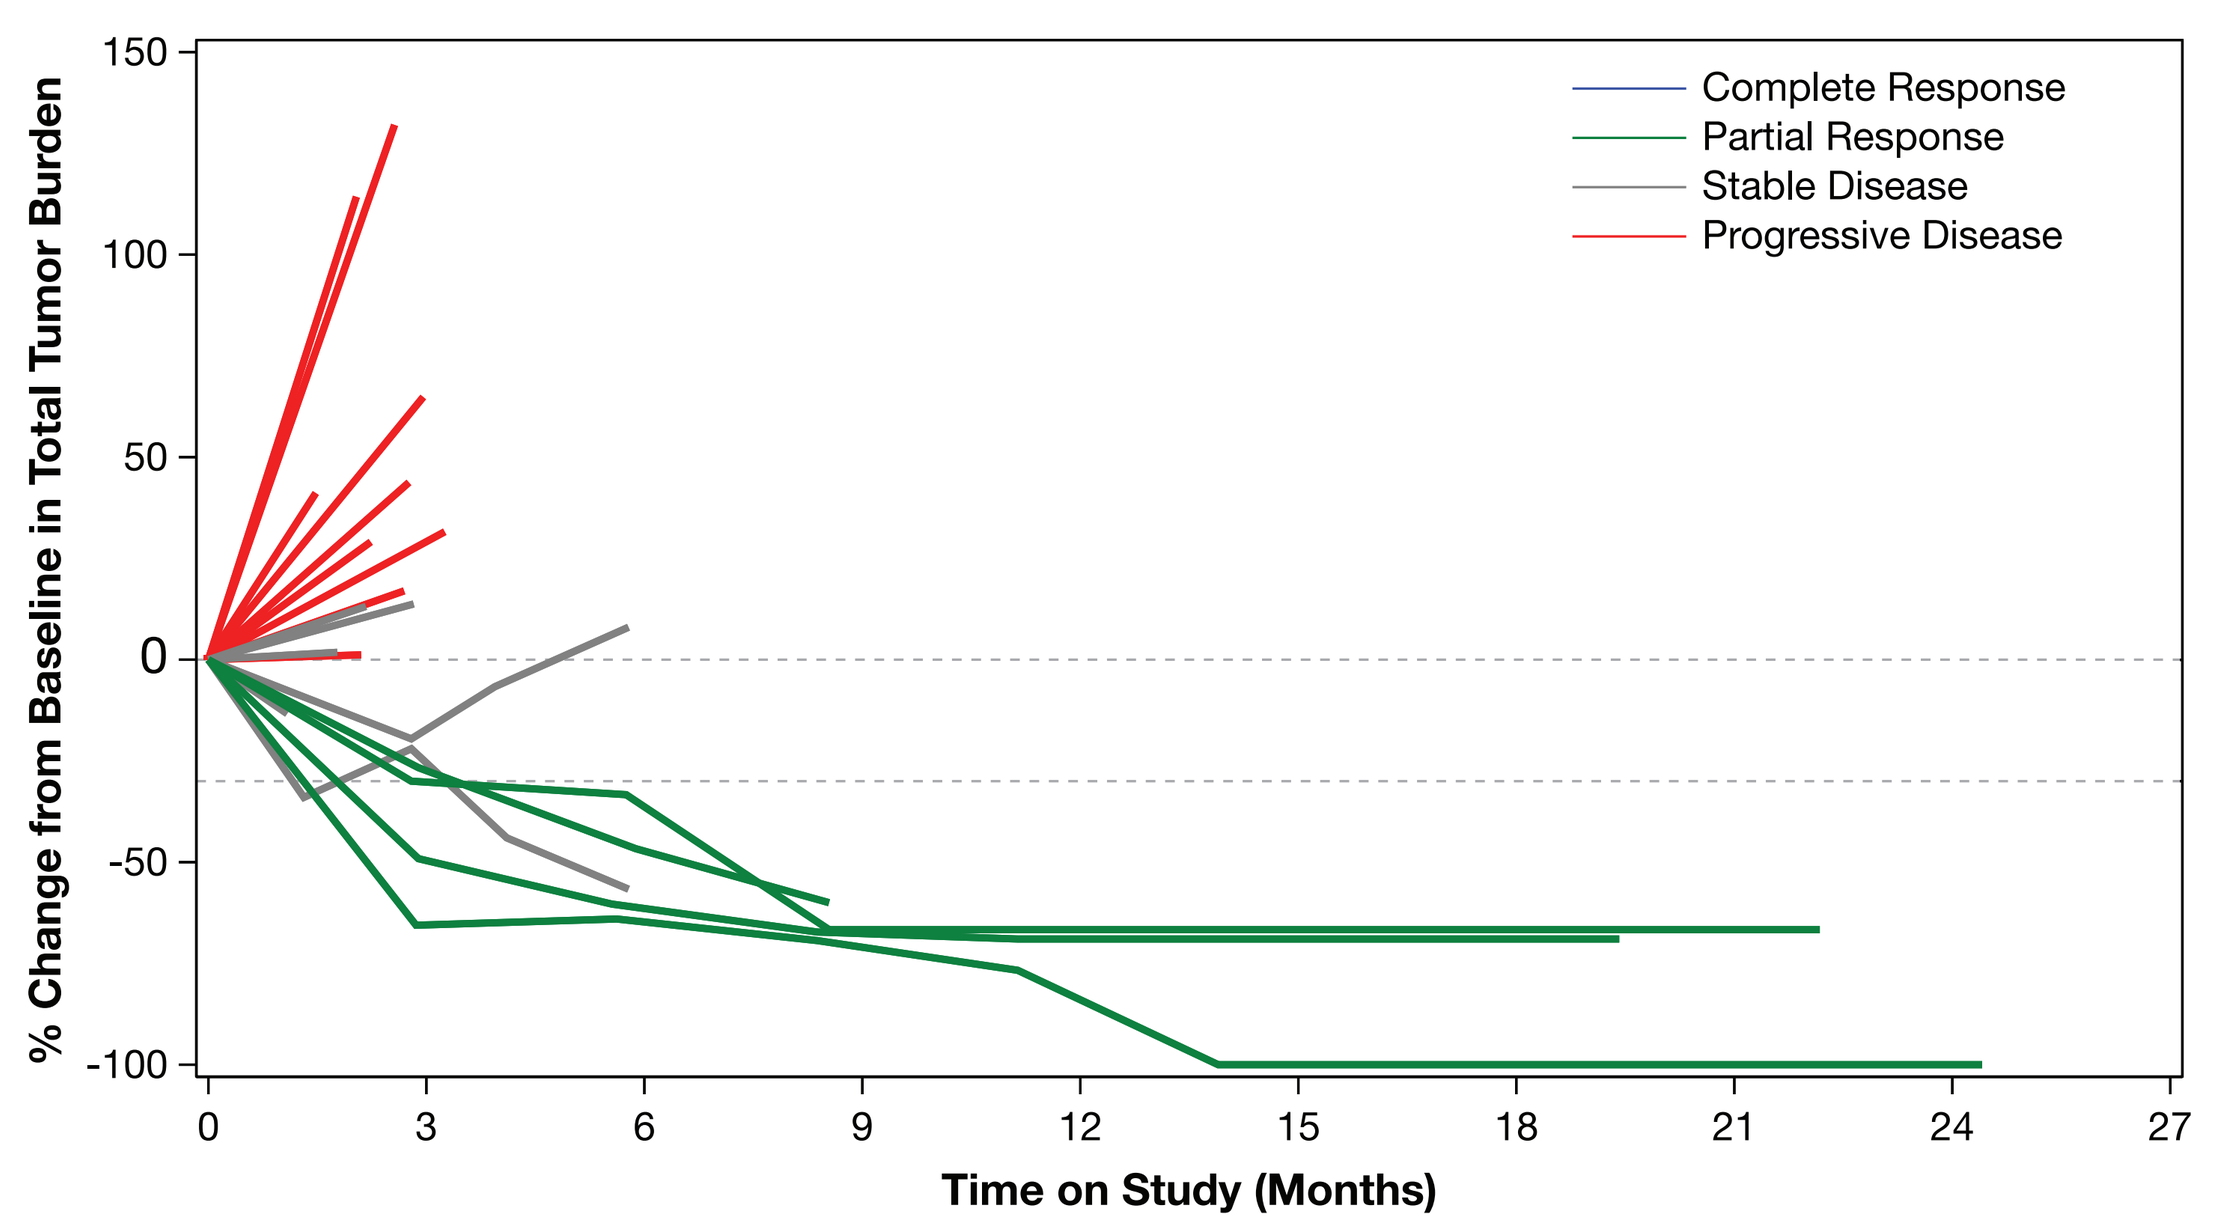
% Change from Baseline in Total Tumor Burden
Complete Response
Partial Response
Stable Disease
Progressive Disease
Time on Study (Months)

Duration of Treatment and Survival Follow-up

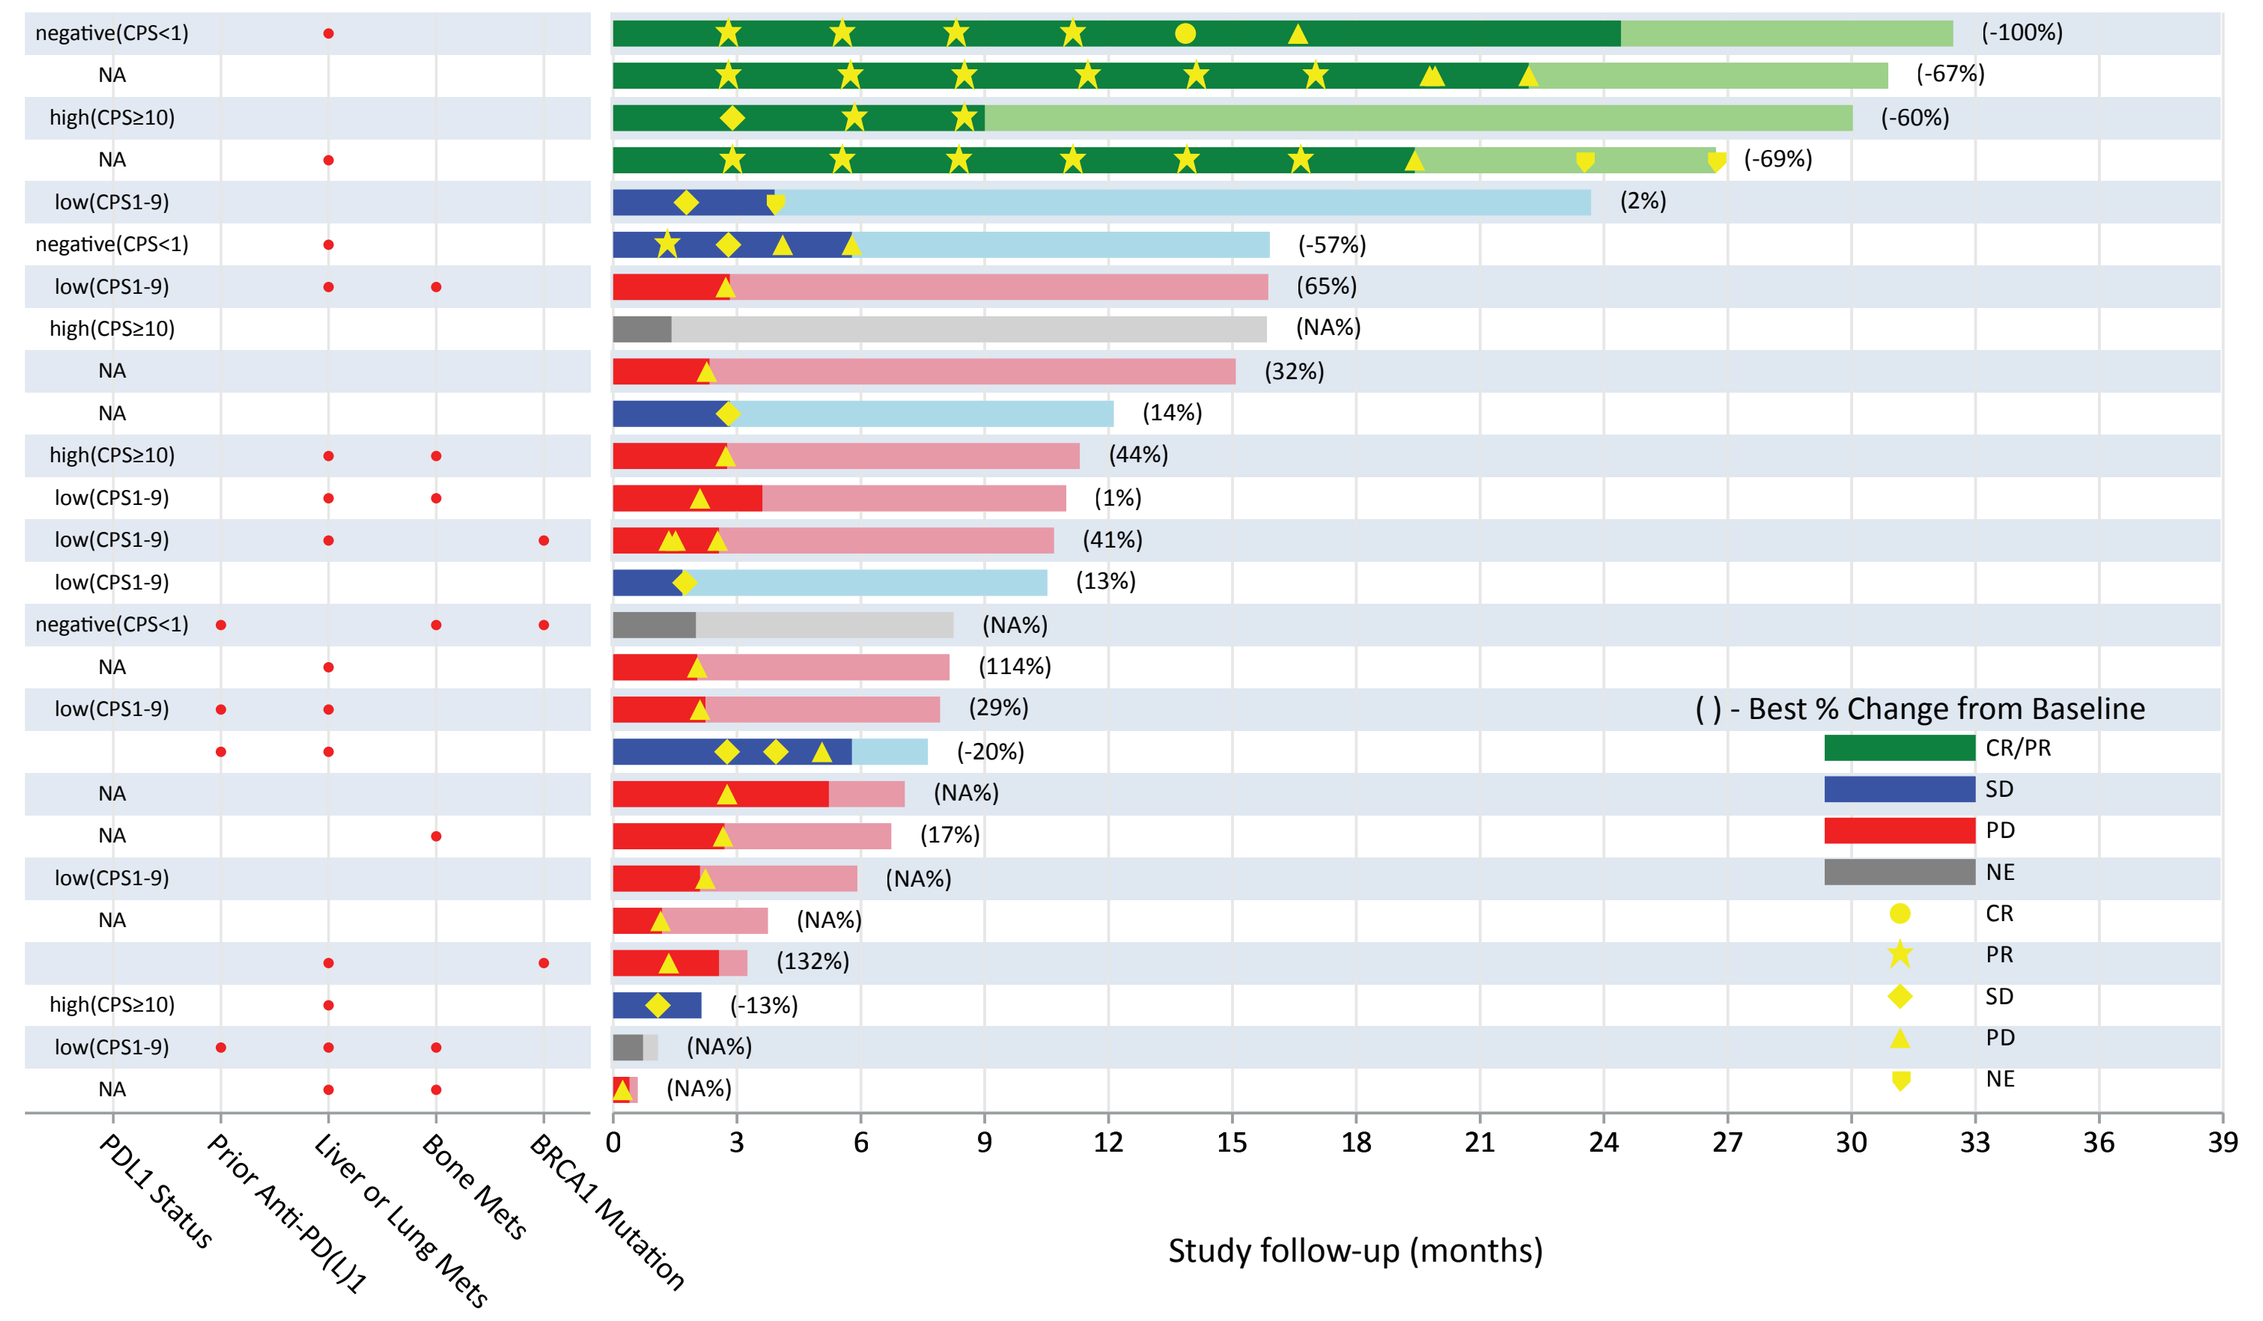
PDL1 Status
Prior Anti-PD(L)1
Liver or Lung Mets
Bone Mets
BRCA1 Mutation
() - Best % Change from Baseline
CR/PR
SD
PD
NE
CR
PR
SD
PD
NE
Study follow-up (months)

KEYNOTE 890: Efficacy Summary (Oct 2021)

Efficacy outcome	Median (95% CI), months
Duration of response	16.6 (13.8–17.0)
Progression-free survival	2.7 (2.1–8.2)
Overall survival	11.0 (7.6–15.9)

Duration of Response

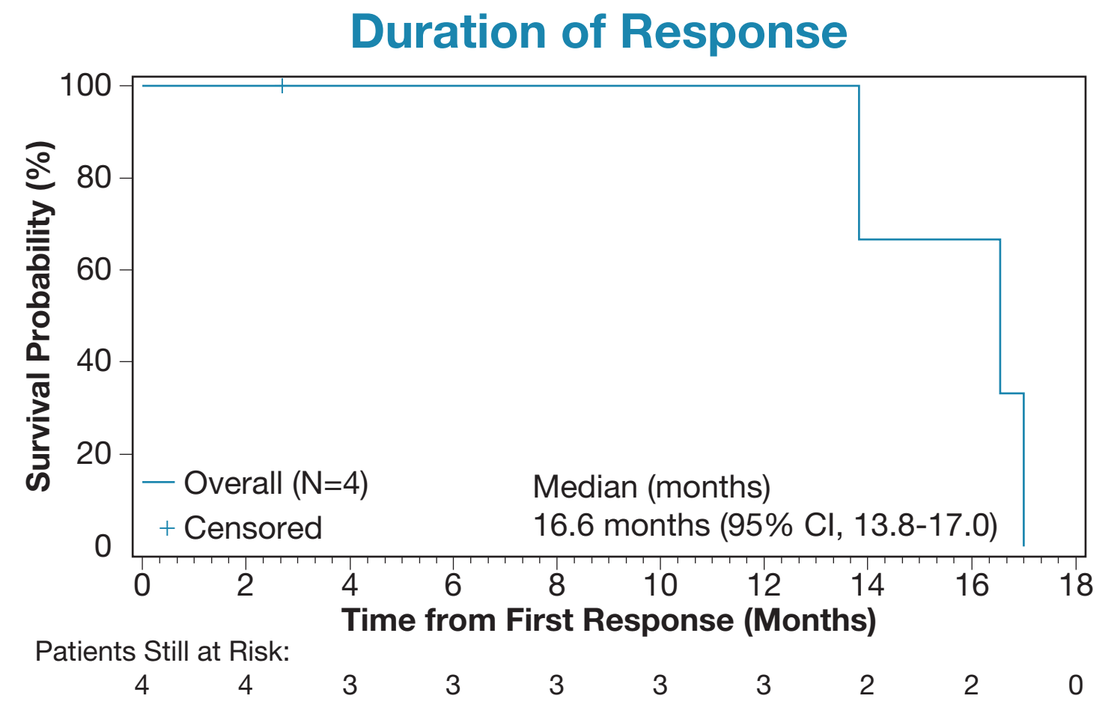
Survival Probability (%)
Overall (N=4)
Censored
Median (months)
16.6 months (95% CI, 13.8-17.0)
Time from First Response (Months)
Patients Still at Risk:
4 4 3 3 3 3 3 2 2 0

Overall Survival

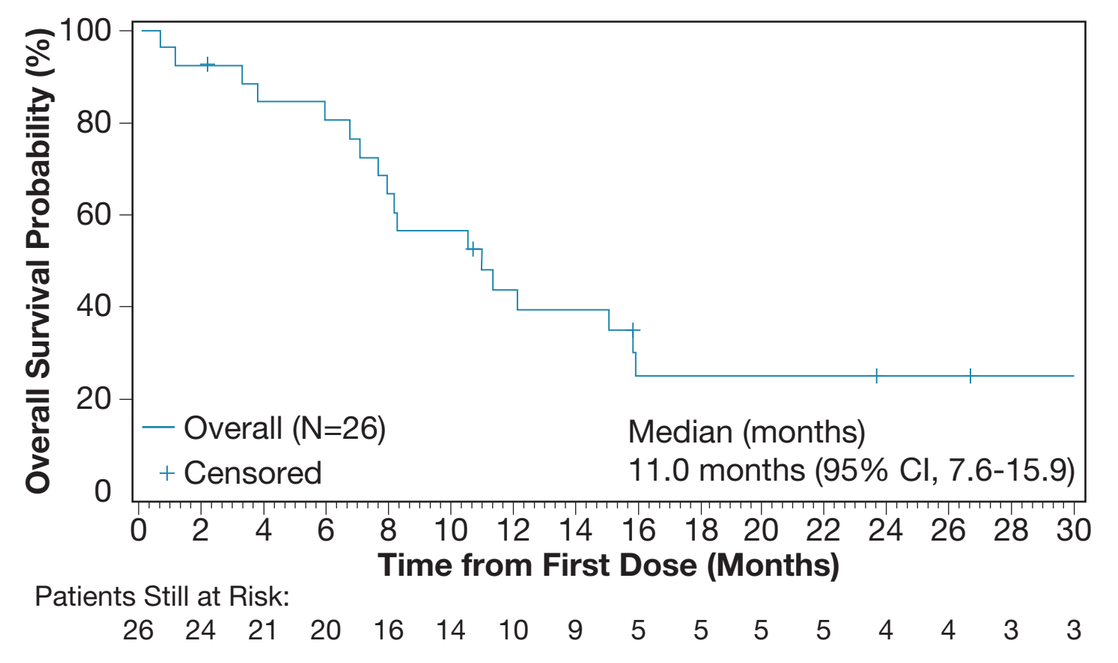
Overall Survival Probability (%)
Overall (N=26)
Censored
Median (months)
11.0 months (95% CI, 7.6-15.9)
Time from First Dose (Months)
Patients Still at Risk:
26 24 21 20 16 14 10 9 5 5 5 5 4 4 3 3

Preliminary, unpublished data.

PATIENT CASE 1
Metastatic TNBC BOR: PR (-69%)

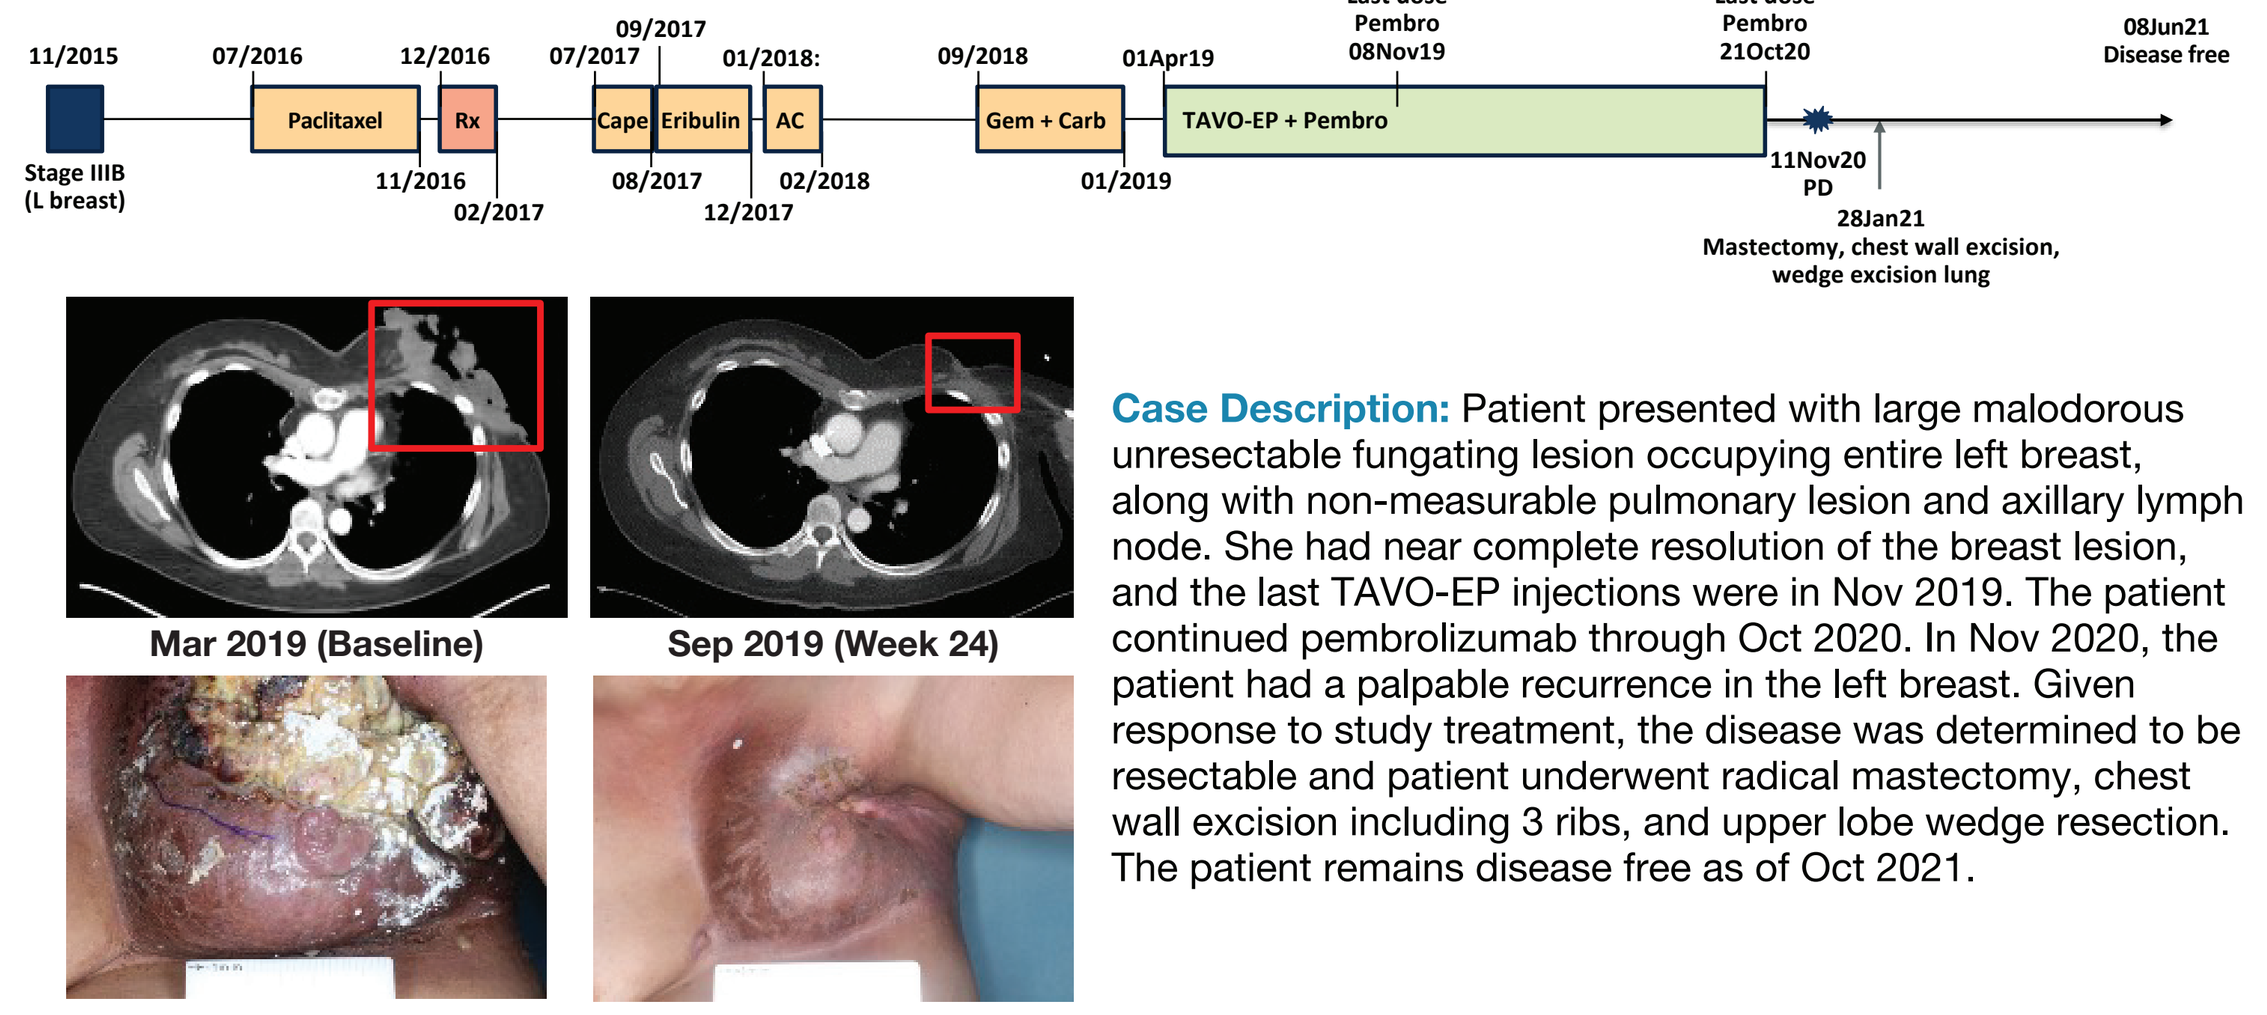
11/2015
Stage IIIB (L breast)
07/2016
Paclitaxel
12/2016
Rx
11/2016
02/2017
07/2017
09/2017
Cape
Eribulin
08/2017
01/2018:
AC
02/2018
12/2017
09/2018
Gem + Carb
01/2019
01Apr19
TAVO-EP + Pembro
Last dose Pembro 08Nov19
Last dose Pembro 21Oct20
11Nov20 PD
28Jan21 Mastectomy, chest wall excision, wedge excision lung
08Jun21 Disease free
Mar 2019 (Baseline)
Sep 2019 (Week 24)

Case Description: Patient presented with large malodorous unresectable fungating lesion occupying entire left breast, along with non-measurable pulmonary lesion and axillary lymph node. She had near complete resolution of the breast lesion, and the last TAVO-EP injections were in Nov 2019. The patient continued pembrolizumab through Oct 2020. In Nov 2020, the patient had a palpable recurrence in the left breast. Given response to study treatment, the disease was determined to be resectable and patient underwent radical mastectomy, chest wall excision including 3 ribs, and upper lobe wedge resection. The patient remains disease free as of Oct 2021.

PATIENT CASE 2
Metastatic TNBC BOR: PR/iCR (-100%)

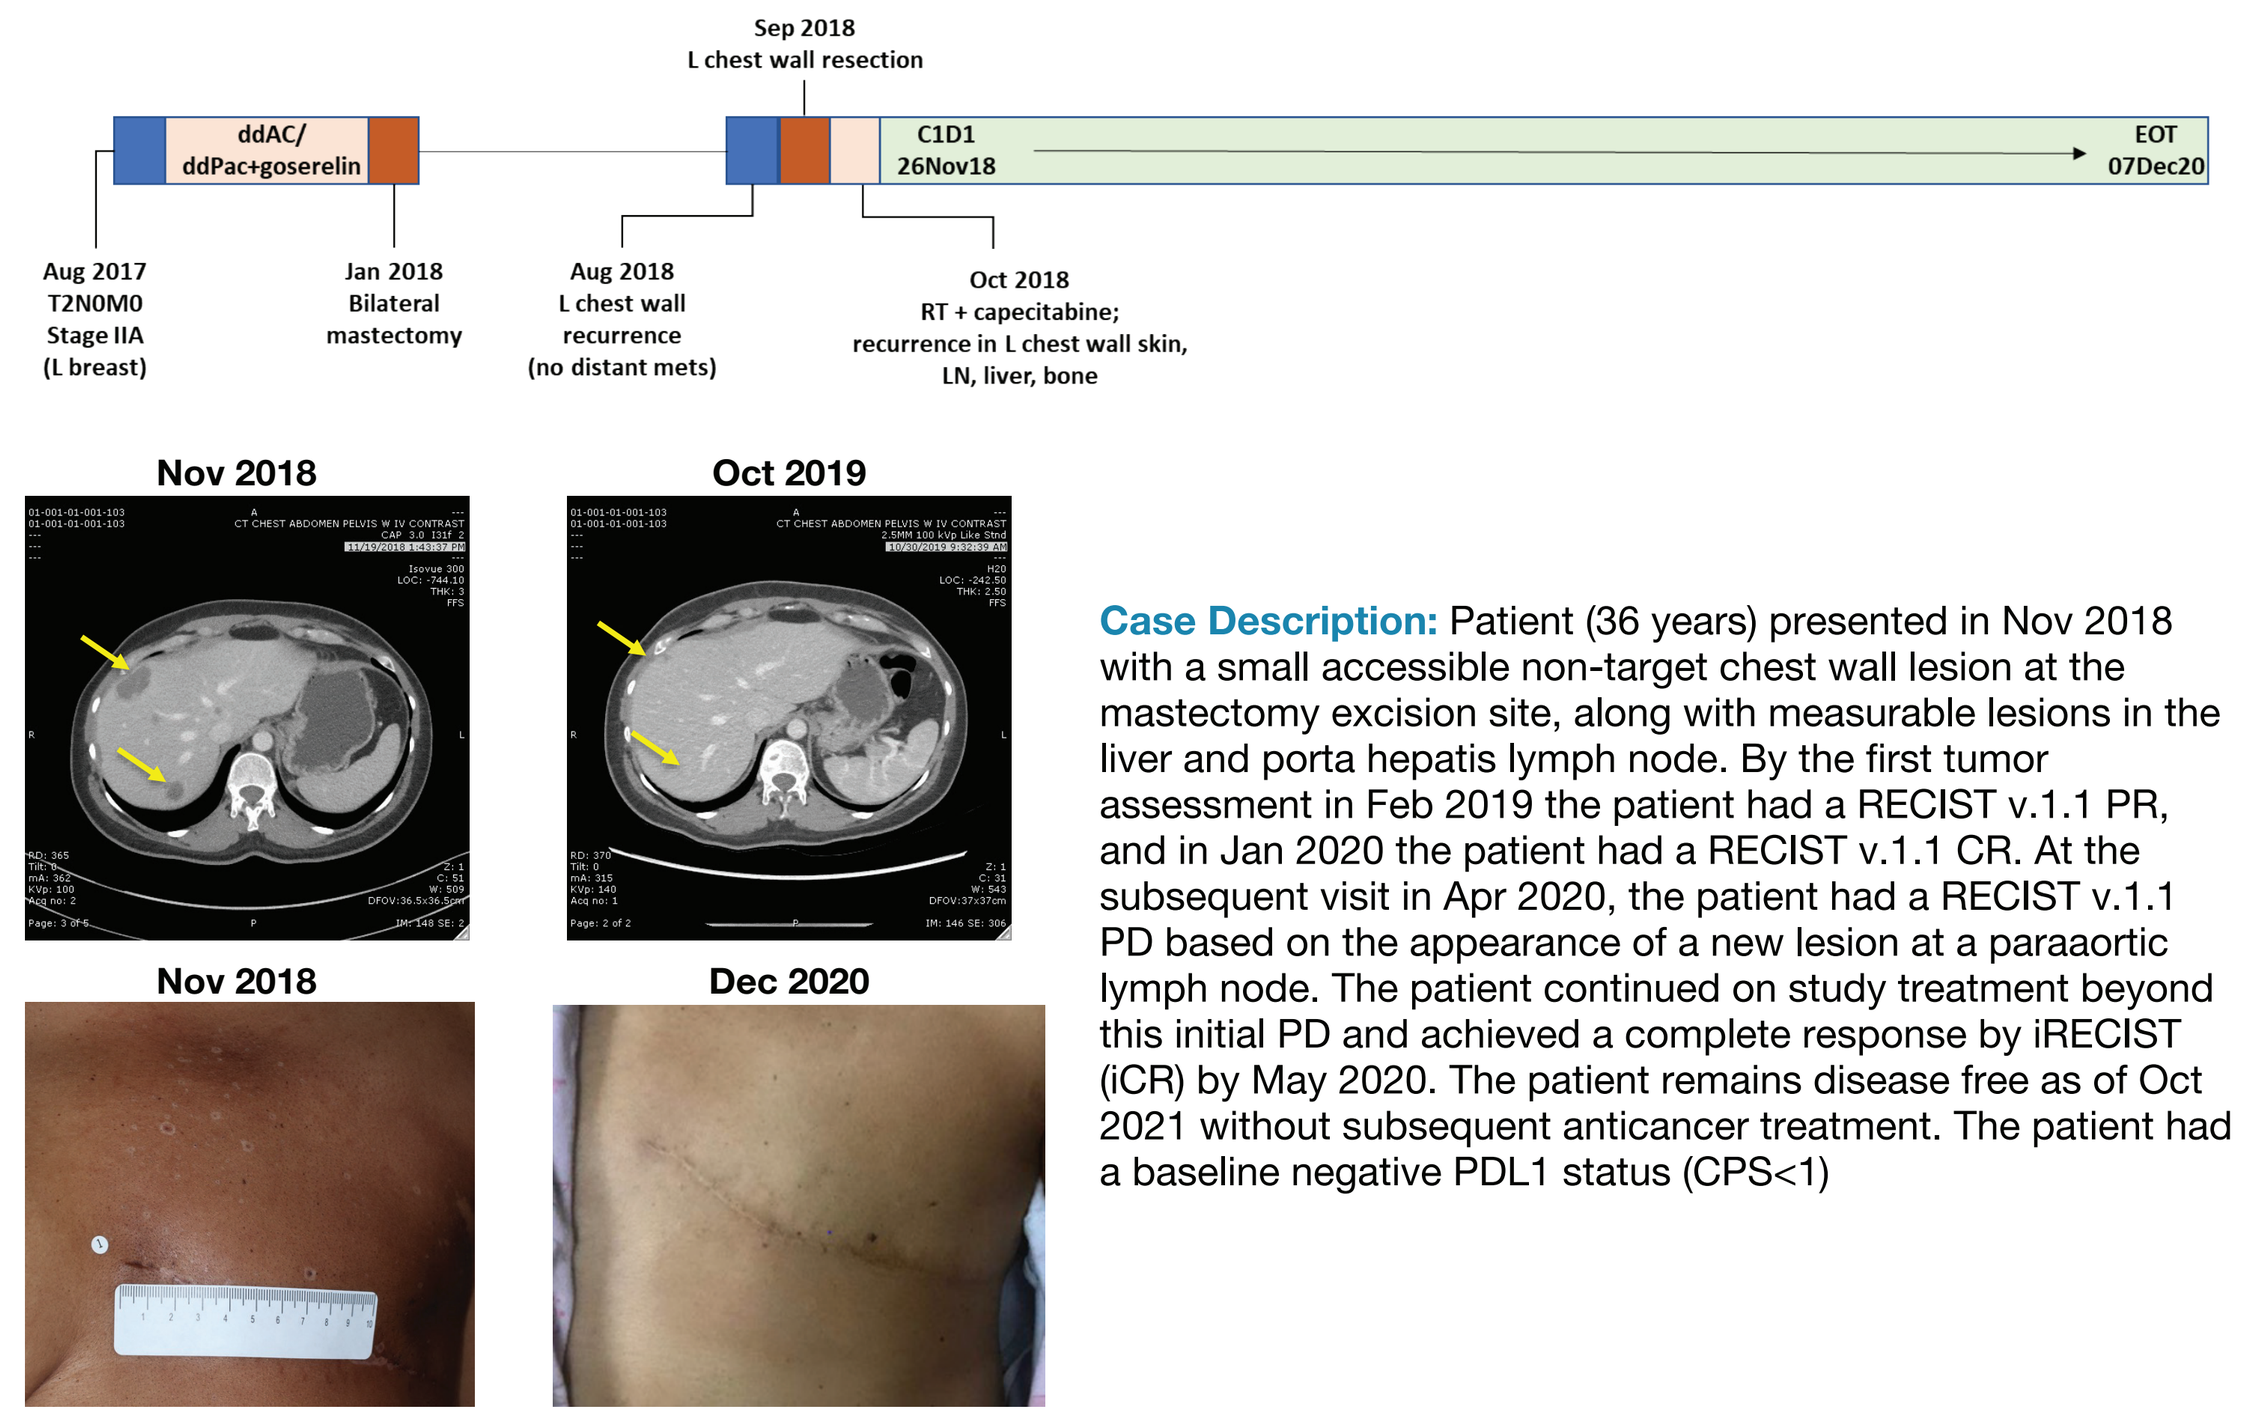
Aug 2017 T2N0M0 Stage IIA (L breast)
ddAC/ ddPac+goserelin
Jan 2018 Bilateral mastectomy
Aug 2018 L chest wall recurrence (no distant mets)
Sep 2018 L chest wall resection
Oct 2018 RT + capecitabine; recurrence in L chest wall skin, LN, liver, bone
C1D1 26Nov18
EOT 07Dec20
Nov 2018
Oct 2019
Nov 2018
Dec 2020

Case Description: Patient (36 years) presented in Nov 2018 with a small accessible non-target chest wall lesion at the mastectomy excision site, along with measurable lesions in the liver and porta hepatis lymph node. By the first tumor assessment in Feb 2019 the patient had a RECIST v.1.1 PR, and in Jan 2020 the patient had a RECIST v.1.1 CR. At the subsequent visit in Apr 2020, the patient had a RECIST v.1.1 PD based on the appearance of a new lesion at a paraaortic lymph node. The patient continued on study treatment beyond this initial PD and achieved a complete response by iRECIST (iCR) by May 2020. The patient remains disease free as of Oct 2021 without subsequent anticancer treatment. The patient had a baseline negative PDL1 status (CPS<1)

PATIENT CASE 3
Metastatic TNBC BOR: SD (-57%)

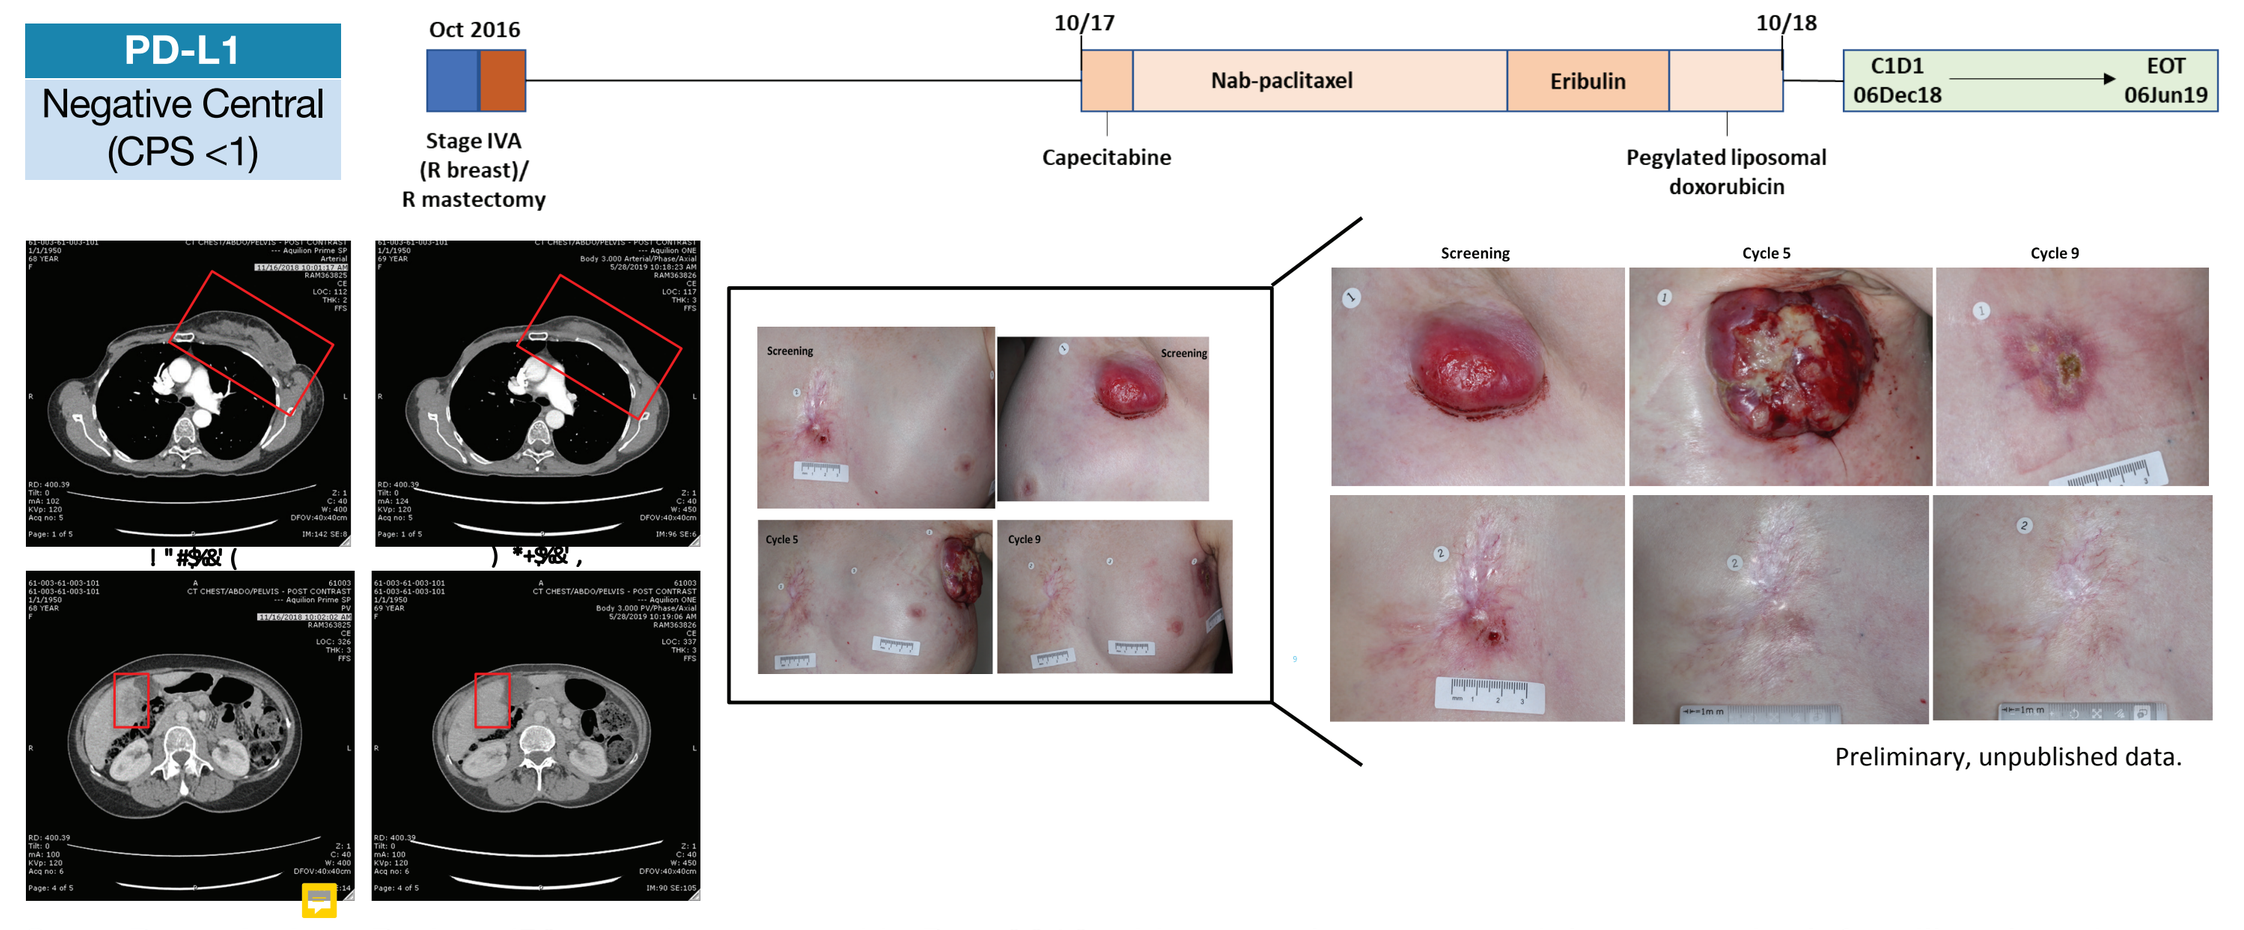
PD-L1
Negative Central (CPS <1)
Oct 2016
Stage IVA (R breast)/ R mastectomy
10/17
Capecitabine
Nab-paclitaxel
Eribulin
10/18
Pegylated liposomal doxorubicin
C1D1 06Dec18
EOT 06Jun19
Screening
Cycle 5
Cycle 9

Preliminary, unpublished data.

Case Description: Patient (52 years) presented in Dec 2018 with a conglomerate nodal mass at the left axillary lymph node and non-target skin involved chest wall lesion over the sternum, in addition to target and non-target liver lesions. The liver and chest wall lesions had reduced in size by the first tumor assessment in Feb 2019, whereas the axillary lymph node lesion appeared to have increased inflammation. By May 2019 the baseline chest and lymph node lesions had nearly resolved and measurable liver lesions reduced in size and met RECIST v.1.1 target lesion response of PR. However the patient had an overall RECIST v.1.1 PD due to the appearance of new non-target liver lesions and the patient discontinued study treatment. The patient had a baseline negative PDL1 status (CPS<1).

TAVO-EP + Pembrolizumab Leads to Productive Immunological Changes in the Periphery

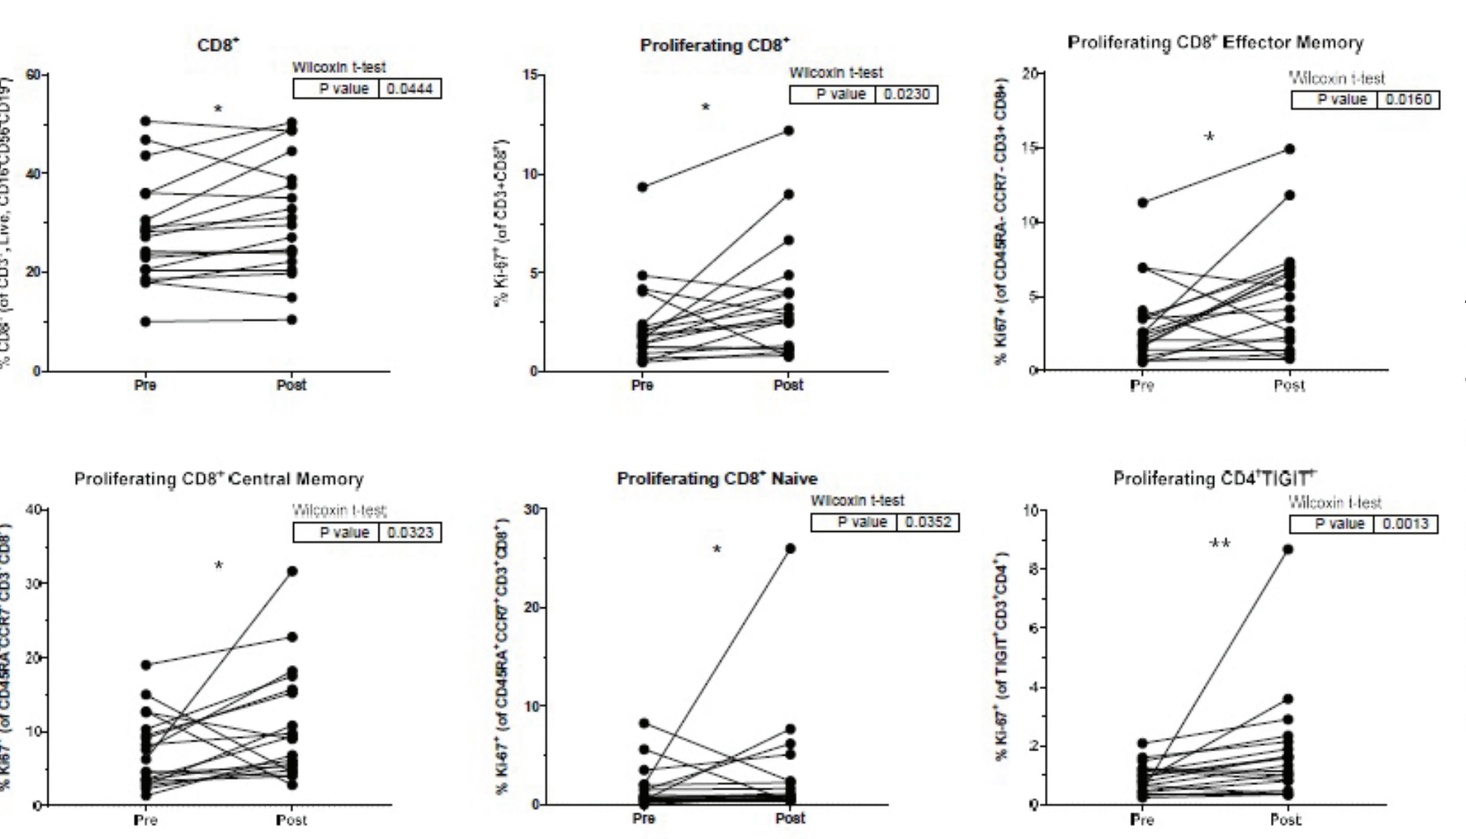
CD8+
Proliferating CD8+
Proliferating CD8+ Effector Memory
Proliferating CD8+ Central Memory
Proliferating CD8+ Naive
Proliferating CD4+TIGIT+
Wilcoxon t-test
P value 0.0444
P value 0.0230
P value 0.0160
P value 0.0323
P value 0.0352
P value 0.0013
Pre
Post

Productive Immunological Changes in the Periphery
Analysis of peripheral immune subsets at screening ("Pre") and post-treatment ("Post") for all evaluable patients shows significant increases in proliferating T cells. Immune subsets shown are CD8+, proliferating CD8+, proliferating CD8+ effector memory, proliferating CD8+ central memory, proliferating CD8+ naïve cells, and proliferating CD4+TIGIT+ cells (n=19 pre/post matched patients with Wilcoxon T tests).

Conclusions

- The combination of plasmid IL-12 (TAVO-EP) and pembrolizumab in pretreated patients with advanced TNBC resulted in durable RECIST v1.1 responses (median 16.6 months).
- Tumor responses were observed in patients with PDL1-negative disease (CPS<1)
- Following treatment on this study, 2 patients have achieved disease-free status which continues >24 months and >30 months after the initial dose of study treatment.
- Substantial reduction of refractory chest wall and other cutaneous lesions impacting patient quality of life was observed in multiple patients.
- Responses in visceral lesions, including durable responses of liver lesions, occurred in multiple patients.
- Significant changes in peripheral immune subsets were detected with combination therapy, • suggesting a productive immune response extending beyond local injected lesions.
- TAVO-EP in combination with pembrolizumab had a mild toxicity profile.
 - The most common treatment-related adverse events (TRAEs) (all grades) were administration site pain and fatigue.
 - Grade 3 TRAEs were reported in 6 patients (23%): fatigue (11.5%); acute kidney injury, enterocolitis, and myocarditis (3.8% each).
 - There were no Grade 4 or 5 TRAEs.
- This novel immunotherapeutic regimen warrants further evaluation in 2L+ advanced TNBC.
- Cohort 2 exploring TAVO-EP + pembrolizumab + chemotherapy in frontline TNBC is currently enrolling (see TiP #OT2-01-03)

References

1. Heller R, *J Clin Oncol.* 2008;26:36.
2. Trinchieri G, *Nat Rev Immunol.* 2003;3:133–146. https://doi.org/10.1038/nri1001
3. Daud, *J Trans Med.* 2015; Algazi, *Clin Cancer Res.* 2020; Algazi, *Ann Onc.* 2020; Telli, *Clin Cancer Res.* 2021.
4. Algazi, *CCR.* 2020; Fernandez-Penas, SITC 2020.
5. Winer, *Lancet Onc.* 2021; Adams *Ann Onc.* 2019; Dirix, *Br Can Res Treat.* 2019.
6. Bardia, *NEJM.* 2021.

This presentation is the intellectual property of the author/presenter. Contact them at (author's email address) for permission to reprint and/or distribute.