PHASE 2, OPEN-LABEL STUDY OF INTRATUMORAL TAVOKINOGENE TELSEPLASMID (TAVO) PLUS ELECTROPORATION IN COMBINATION WITH INTRAVENOUS PEMBROLIZUMAB THERAPY IN PATIENTS WITH INOPERABLE LOCALLY ADVANCED OR METASTATIC TRIPLE-NEGATIVE BREAST CANCER (KEYNOTE-890)

Authors: Melinda L. Telli[1], Irene Wapnir[1], Bianca Devitt[2], Katharine Cuff[3], Hatem Soliman[4], Shaveta Vinayak[5], Alain Algazi[6], Donna Bannavong[7], Erica Browning[7], David A. Canton[7], Christopher Twitty[7], Kellie Malloy Foerter[7] and Rohit Joshi[8]

Affiliations: [1]Stanford University School of Medicine, Stanford, CA, USA, [2]Box Hill Hospital, Box Hill, Vic, AU, [3]Princess Alexandra Hospital, Woolloongabba, QE, AU, [4]Moffit Cancer Center, Tampa, FL, USA, [5]University of Washington, Fred Hutchinson Cancer Research Center, Seattle Cancer Care Alliance, Seattle, WA, USA, [6]University of California San Francisco Helen Diller Cancer Center,[7] OncoSec Medical Incorporated, San Diego, CA and Pennington, NJ [8], Adelaide Oncology and Haematology, Adelaide, AU.

ACKNOWLEDGEMENTS
Special thanks to the study participants, their families, the institutions participating in KEYNOTE 890 and to Susan G. Komen for their support of aspects of this work.

Background

Emerging studies show that TNBC tumors with a proinflammatory microenvironment are associated with better outcomes[4-6], immune-modulating therapies, like anti-PD-1 and anti-PD-L1 monoclonal antibodies, have demonstrated modest activity in pre-treated subjects with metastatic TNBC, with objective response rates (ORR) <10%[2]. KEYNOTE-086 showed an ORR of 5.3% and KEYNOTE-119 showed an ORR of 9.6%[10,11]. Local intratumoral administration of tavokinogene telseplasmid (plasmid encoding interleukin-12) followed by electroporation (TAVO™) of accessible lesions is hypothesized to enhance tumor immunogenicity.

A phase 1 study in subjects with locally advanced or recurrent TNBC showed that TAVO™ is a safe and tolerable therapy for skin and subcutaneous TNBC tumors[7]. Combining TAVO™ with an anti-PD-1 antibody, such as pembrolizumab, may improve responses in subjects with mTNBC by potentially converting poorly-immunogenic/low tumor-infiltrating lymphocyte (TIL) tumors into immune-responsive/high TIL tumors.

Mechanism of Action of IT-tavo-EP (TAVO™)

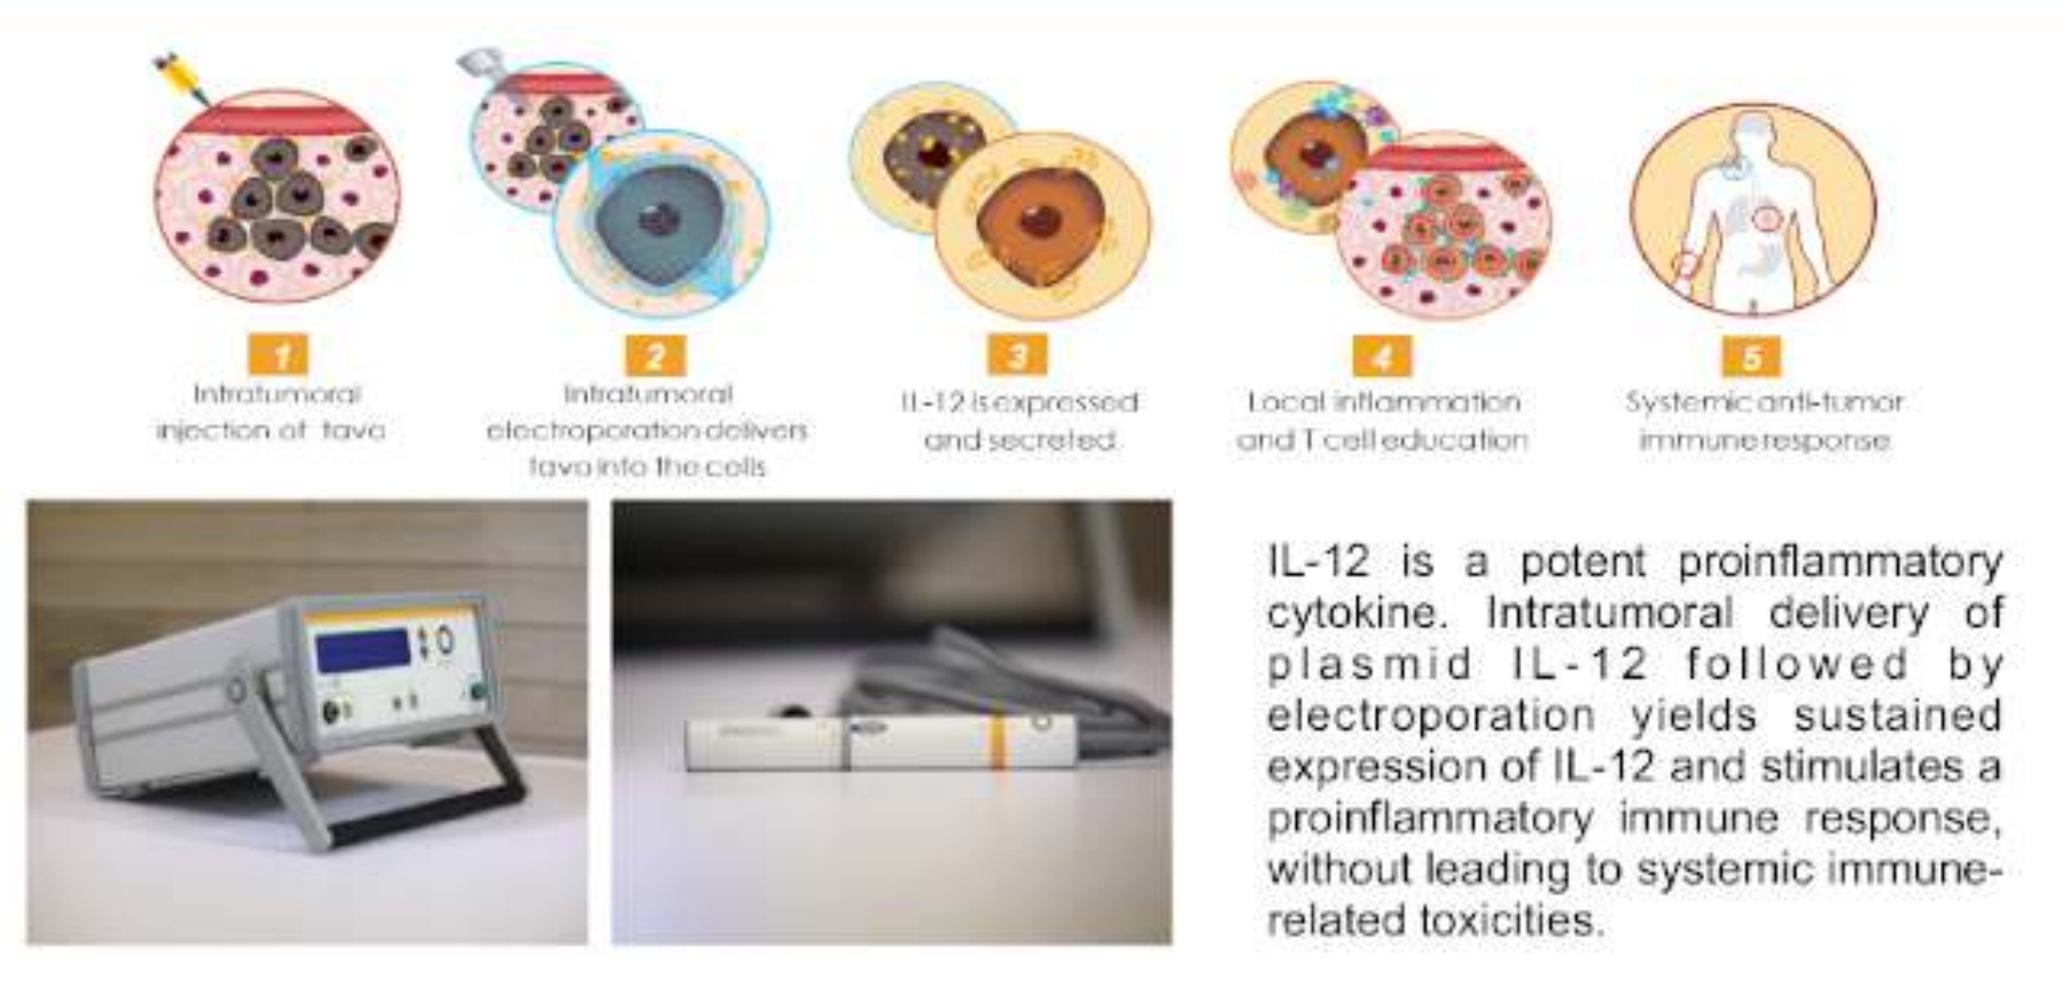
1 Intratumoral injection of tavo
2 Intratumoral electroporation delivers tavo into the cells
3 IL-12 is expressed and secreted
4 Local inflammation and T cell education
5 Systemic anti-tumor immune response

IL-12 is a potent proinflammatory cytokine. Intratumoral delivery of plasmid IL-12 followed by electroporation yields sustained expression of IL-12 and stimulates a proinflammatory immune response, without leading to systemic immune-related toxicities.

KEYNOTE-890 Trial Design

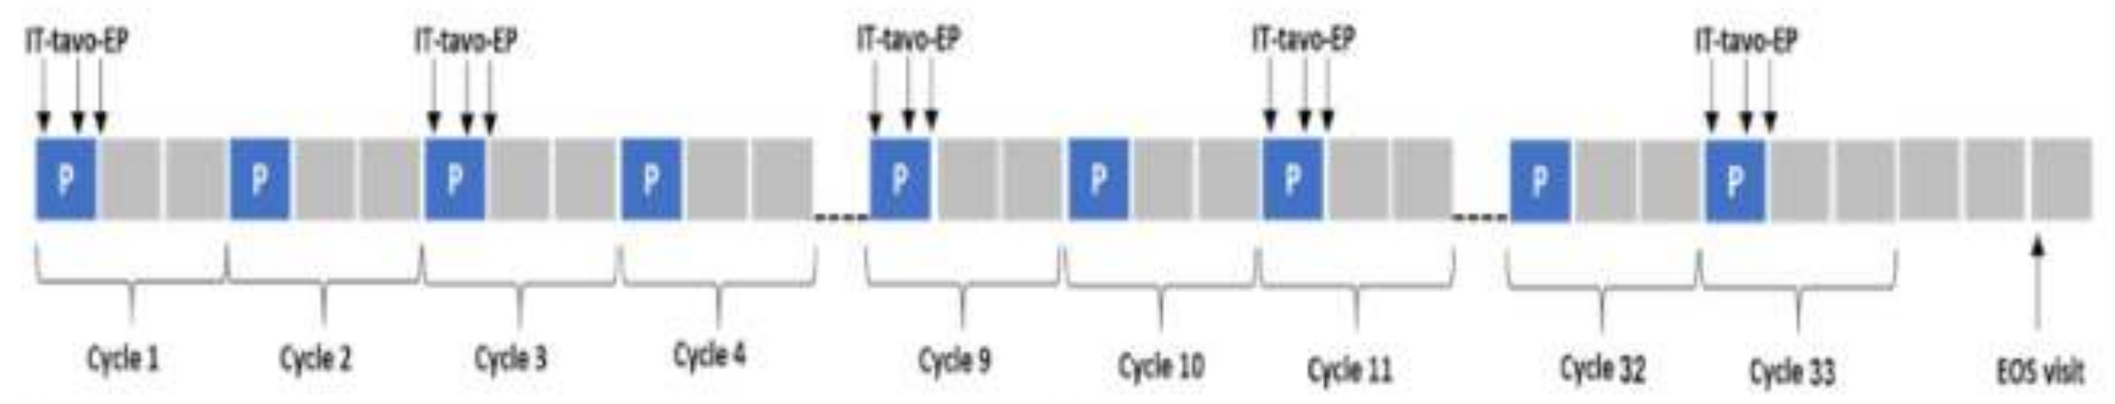
IT-tavo-EP
P
Cycle 1
Cycle 2
Cycle 3
Cycle 4
Cycle 9
Cycle 10
Cycle 11
Cycle 32
Cycle 33
EOS visit

- Single-arm, phase II study (NCT03567720) of TAVO™ + pembrolizumab in mTNBC.
- Subjects are treated with 0.5 mg/mL TAVO™ to accessible lesions on days 1, 5, and 8 every 6 weeks, and with 200 mg IV pembrolizumab on day 1 of each 3-week cycle for up to 33 cycles pembrolizumab. TAVO™ is injected at a dose volume of ~¼ of the calculated lesion volume, with a minimum dose of 0.1 mL. Six pulses at a field strength of 1500 volt/cm and pulse width of 100 µs at 300-msec intervals are delivered using a handheld electroporation device.
- Imaging for RECIST v1.1 assessment is completed every 12 weeks.

Eligibility Criteria

- Histologically confirmed TNBC and at least 1 prior line of systemic chemotherapy or immunotherapy, including anti-PD1/PD-L1.
- ER and PR staining <10% and HER2-negative defined as IHC 0 to 1+ or FISH negative.
- RECIST v1.1 measurable disease and at least one anatomically distinct lesion accessible for intratumoral injection and electroporation.
- Subjects must be ≥18 years with ECOG performance status of 0-1.
- Life expectancy of at least 6 months.
- Disease not amenable to potentially curative treatment.

KEYNOTE-890 Endpoints

Primary Endpoint
- Objective Response Rate (ORR) by investigator review based on RECIST v1.1

Secondary Endpoint
- Safety and tolerability of the combined therapy
- Duration of response (DOR), immune ORR, progression-free survival (PFS), immune PFS, and overall survival (OS) of the combined therapy

Exploratory Endpoint
- To characterize the local and systemic immunological changes induced by treatment and to compare these changes in responders and non-responders

Statistical Analysis

- Twenty-Five (25) patients are included in this open-label, single arm, signal finding trial to determine the magnitude of effect for TAVO™ in combination with pembrolizumab on ORR in pre-treated mTNBC prior to the advancement to a larger, more definitive efficacy trial.
- Disease response is assessed using RECIST v1.1. The ORR and iORR will be accompanied by a 2-sided 95% exact binomial confidence interval (CI). Time-to-event endpoints such as DOR, PFS, iPFS, and OS will be analyzed using the Kaplan-Meier method.
- Adverse events is assessed using CTCAE version 5.0.

Table 1. Subject Demographic and Baseline Characteristics

Characteristic	Safety Population (N=16)	Evaluable Population [a] (N=14)
Mean age, years (range)	52 (33-70)	52 (33-70)
Female, n (%)	16 (100.0)	14 (100.0)
Race, n (%)		
Black or African American	3 (18.7)	2 (14.3)
Native Hawaiian or Other Pacific Islander	1 (6.3)	1 (7.1)
White	11 (68.7)	10 (71.4)
Other	1 (6.3)	1 (7.1)
PD-L1 Status[b]		
Positive	3 (18.8)	3 (21.4)
Negative	4 (25.0)	4 (28.6)
Unknown	9 (56.2)	7 (50.0)
Median prior lines (range 1 – 6)		
Number of prior lines of systemic therapy, n (%)[c]	3	3
1	3 (18.7)	2 (14.3)
2	4 (25.0)	3 (21.4)
≥3	9 (56.3)	9 (64.3)
Prior (neo)adjuvant therapy	13 (81.3)	11 (78.7)

a. Completed at least 1 radiographic assessment or discontinued prior to assessment due to clinical disease progression.
b. PD-L1 status was provided where available but was not required for trial.
c. Prior lines including adjuvant and neoadjuvant treatments.

Table 2. Efficacy in Evaluable Population

Table 2. Investigator RECIST v1.1 Best Overall Response

		(N=14)
Objective Response Rate (ORR) = (CR +PR)		**4 (28.6)**
95% Exact Binomial Lower Limit		**(8.39, 58.10)**
Best Overall Response: n (%)	Complete Response (CR)	0 (0.0)
	Partial Response (PR)	4 (28.6)
	Stable Disease (SD)	3 (21.4)
	Progressive Disease (PD)	7 (50.0)

Table 3. Characteristics of the Evaluable Population

Table 3. Characteristics of the Evaluable Population (N=14)

Characteristic	Responders (N=4)	Non-Responders (N=10)
Prior Therapy for Advanced Disease, n (%)		
Chemotherapy	3 (75.0)	9 (90.0)
Endocrine Therapy	1 (25.0)	1 (10.0)
Anti-PD-L1 Antibody Therapy	0 (0.0)	1 (10.0)
Other Targeted Therapy	0 (0.0)	2 (20.0)
ECOG Performance Status at Enrollment		
0	2 (50.0)	5 (50.0)
1	2 (50.0)	5 (50.0)

REFERENCES

1. Adams S, et al. *J Clin Oncol.* 2014;32(27):2959–66.
2. Loi S, et al. *Cancer Res.* 2016;76(4):S1–03.
3. Loi S, et al. *Ann Oncol.* 2014;25(8):1544–50.
4. 2Dua I, et al. *Am J of Hem Oncol.* 2017;13(4):20–7.
5. American Cancer Society. *Breast Cancer Facts & Figures 2017–2018.* Atlanta: American Cancer Telli ML, et al. *Cancer Res* 2018;78(13 Suppl):Abstract nr. CT022.
6. Rosner S. *Cancer Med.* 2018 Mar;7(3):690-697.
7. Vareki S. *Crit Rev Oncol Hematol.* 2017 Aug;116:116-124.
8. Alexander M. *Med J Aust.* 2014 Jul 7;201(1):49-53.
9. Powles T. *J Clin. Oncol.* 2017;35 (3035-3035).
10. Cortes,J. *Ann. Oncol.* 2019;30 (suppl_5):v851-v934.
11. Adams, S. *Ann Oncol.* 2019; 30 (3):397-404.

Table 4. Safety - Treatment Emergent Adverse Events

Table 4. Treatment Emergent Adverse Events (≥ 10 %)

	Any Grade N (%)	Grade ≥3 N (%)	Grade 5 N (%)
Number of subjects reporting at least one TEAE	16 (100.0)	3 (18.8)	0 (0.0)
Preferred Term			
Administration site pain	11 (68.8)	0 (0.0)	0 (0.0)
Fatigue	5 (31.3)	1 (6.3)	0 (0.0)
Hypothyroidism	4 (25.0)	0 (0.0)	0 (0.0)
Anxiety	2 (12.5)	0 (0.0)	0 (0.0)
Dyspnea	2 (12.5)	0 (0.0)	0 (0.0)
Nausea	2 (12.5)	0 (0.0)	0 (0.0)
Vomiting	2 (12.5)	0 (0.0)	0 (0.0)
Acute kidney injury	1 (6.3)	1 (6.3)	0 (0.0)
Hyperglycemia	1 (6.3)	1 (6.3)	0 (0.0)

Figure 1: Best Overall Response Includes Regression of Tumors Lacking PD-L1 Expression

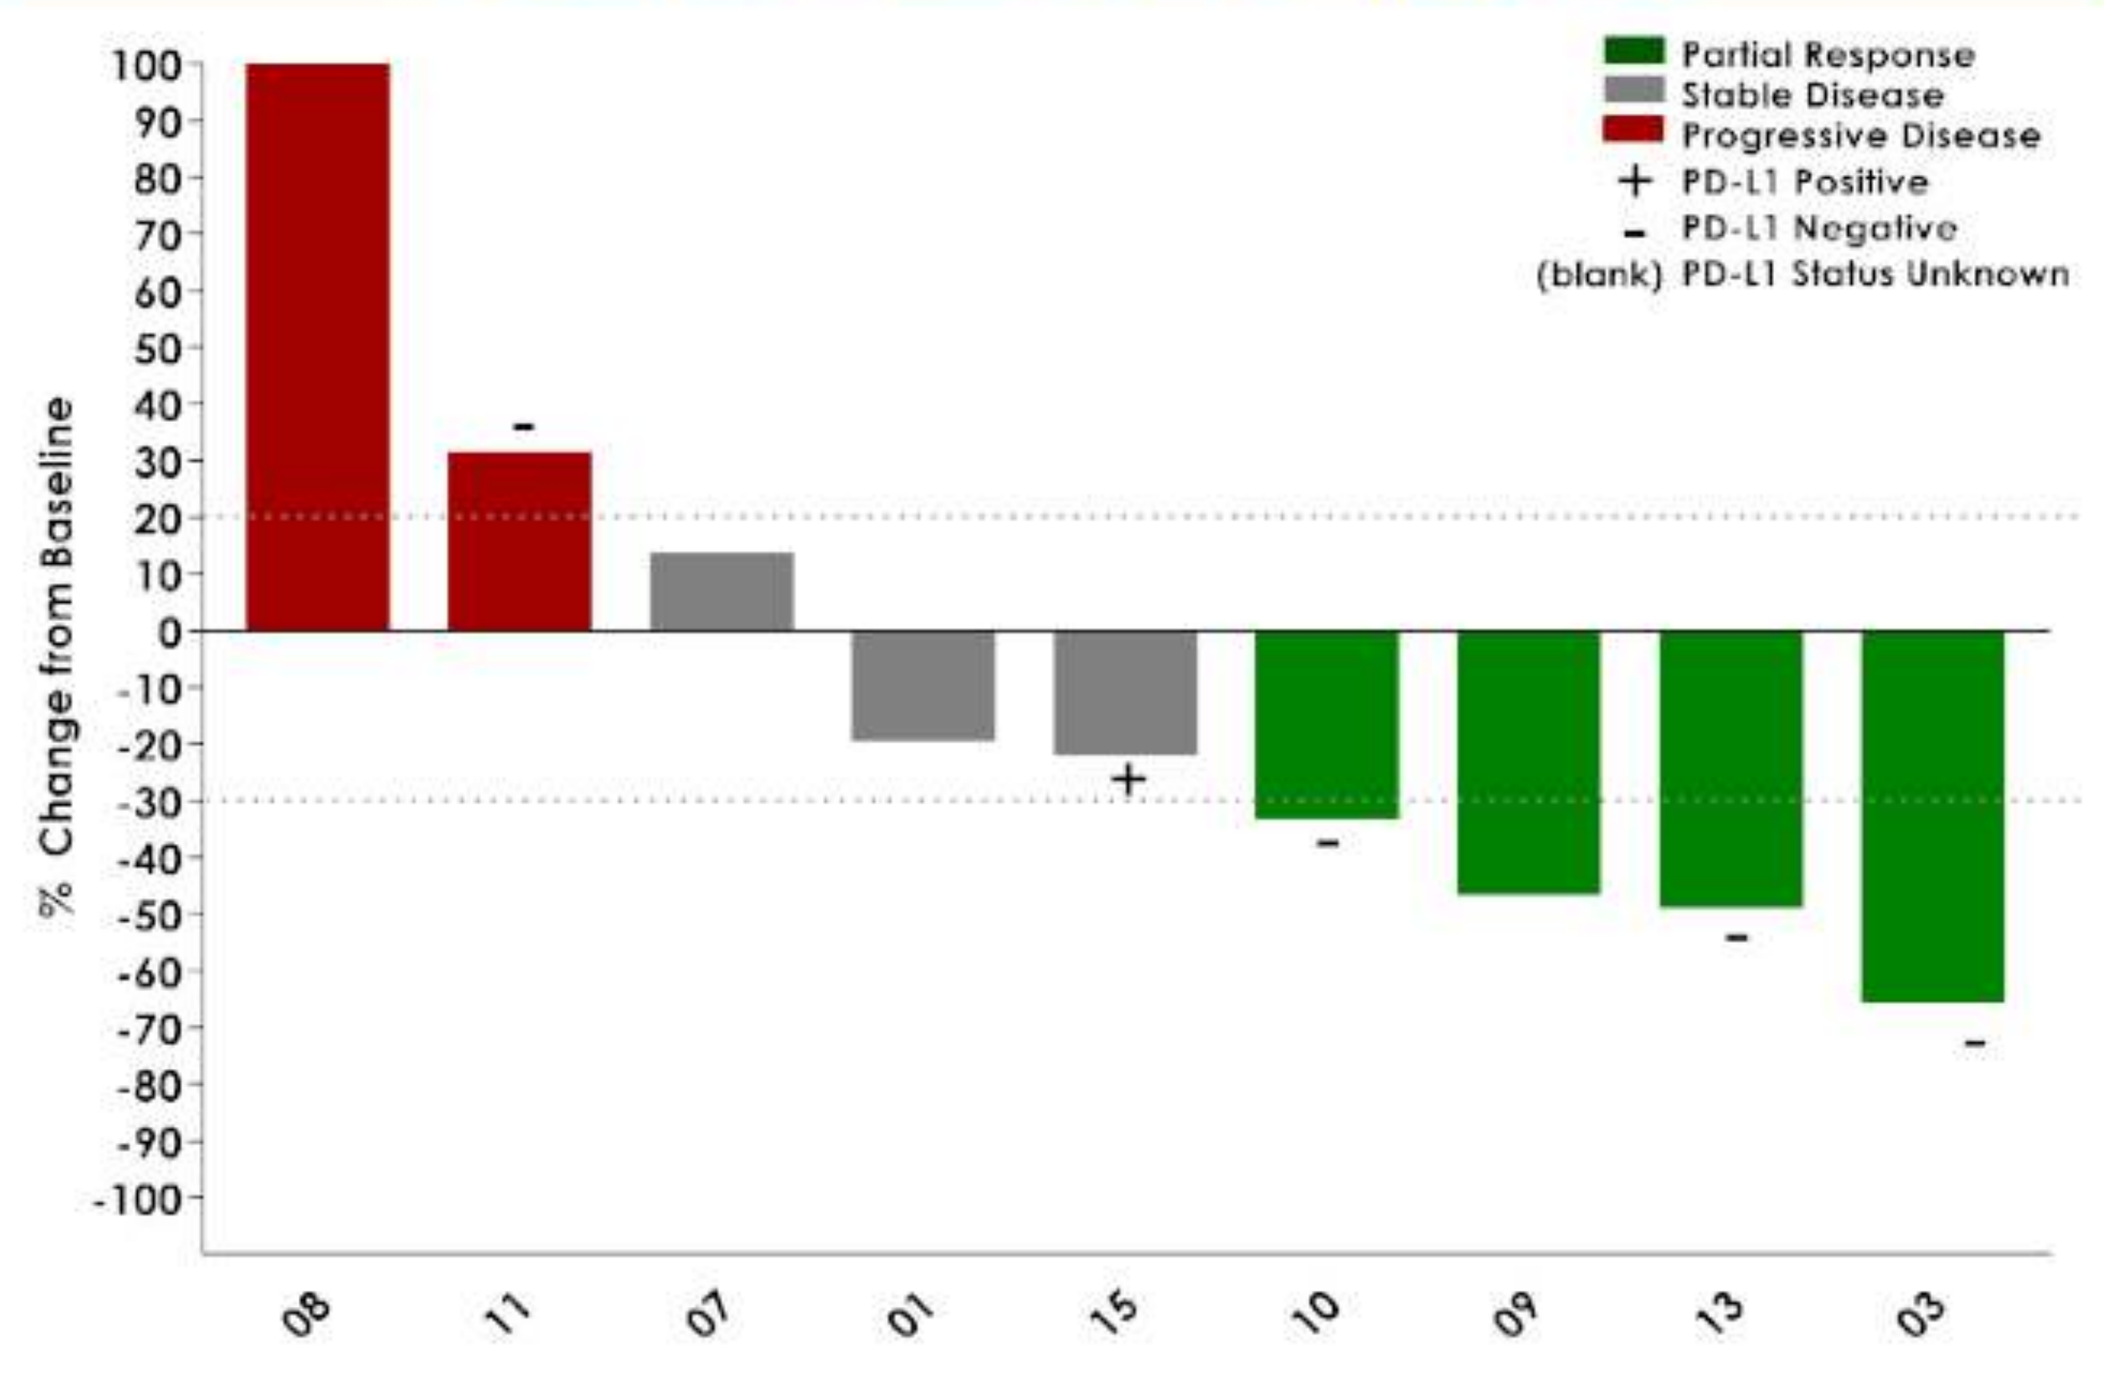
Partial Response
Stable Disease
Progressive Disease
+ PD-L1 Positive
- PD-L1 Negative
(blank) PD-L1 Status Unknown
% Change from Baseline
100
90
80
70
60
50
40
30
20
10
0
-10
-20
-30
-40
-50
-60
-70
-80
-90
-100
08
11
07
01
15
10
09
13
03

Figure 1: Percent change of target lesions at BOR timepoint from baseline.

Note: Five subjects did not have post-treatment measurements and discontinued due to clinical disease progression not graphed. Three patients with PD-L1 negative tumors were assessed as PR.

PD-L1 status assessments are ongoing, available assessments are provided above using screen biopsies or archival testing.

Figure 2: Time to Response and Duration of Response

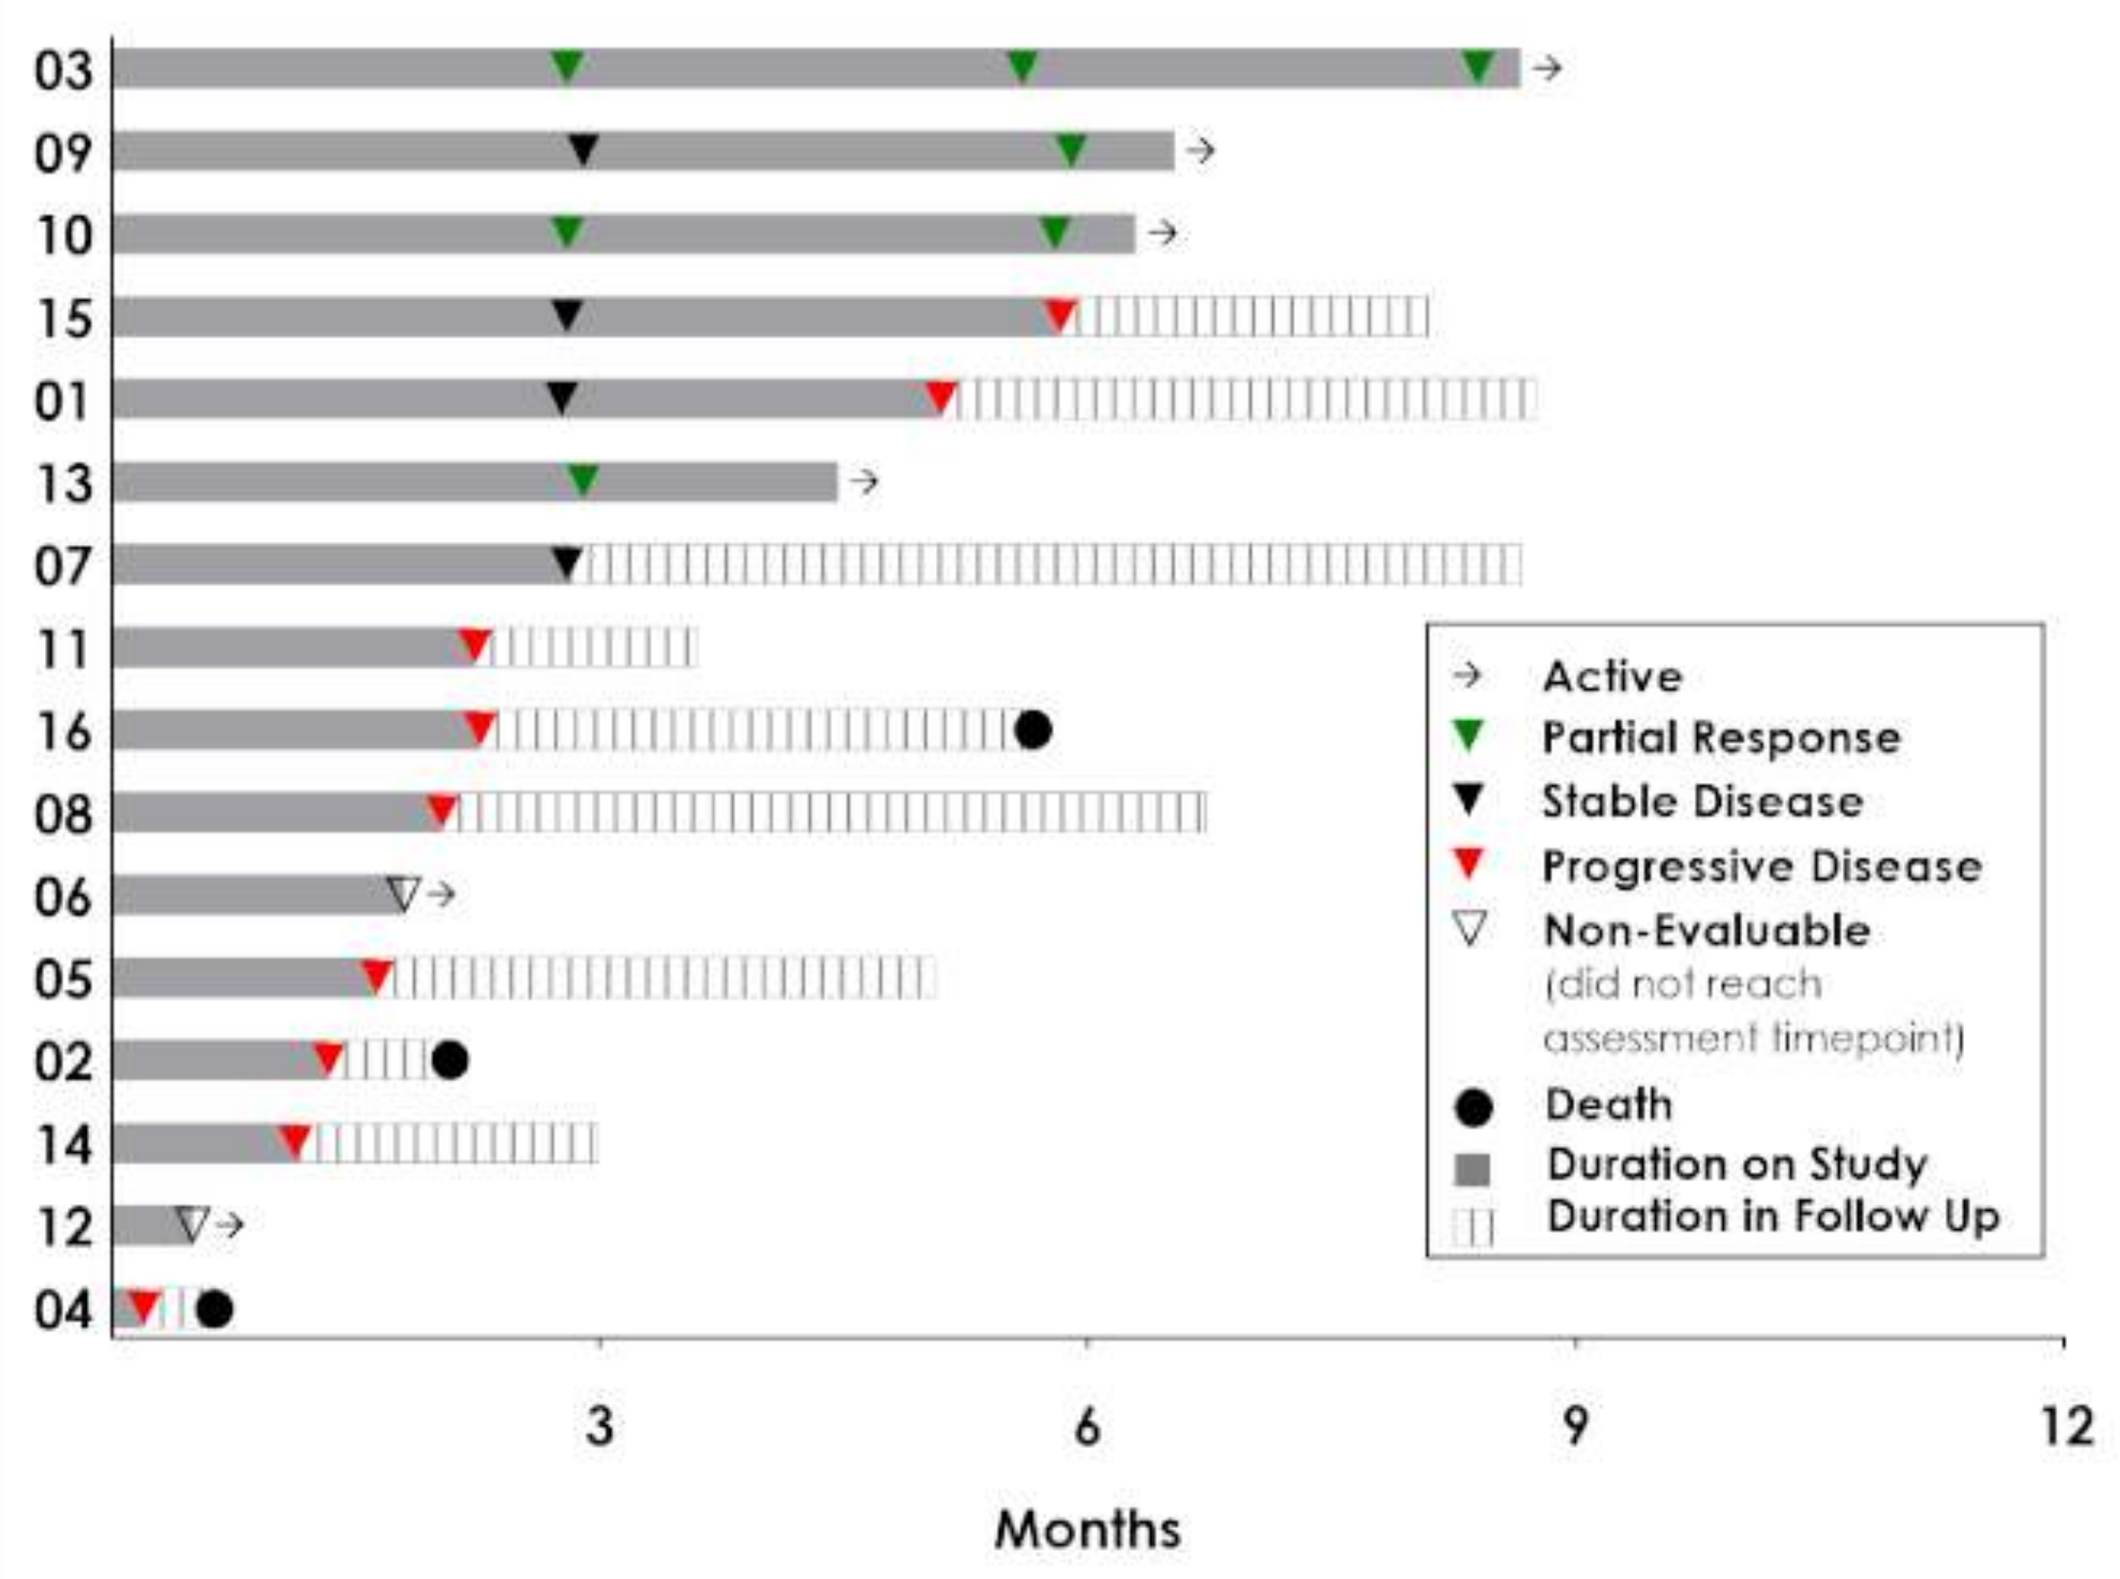
03
09
10
15
01
13
07
11
16
08
06
05
02
14
12
04
Active
Partial Response
Stable Disease
Progressive Disease
Non-Evaluable
(did not reach assessment timepoint)
Death
Duration on Study
Duration in Follow Up
3
6
9
12
Months

Figure 2: Average time to response was 3.6 months. All 4 responders are ongoing as of 13Aug19 data cut.

Figure 3: Images from Responding Patients

Patient # 03

- 35 year-old female
- PD-L1 negative by Ventana SP-142 and Dako 22C3
- Rapid relapse following neoadjuvant chemotherapy and rapid progression on 1st line chemotherapy
- PR by RECIST v1.1 with a 66% reduction in SLD
- Resolution of chest wall disease and regression of distant hepatic and nodal disease

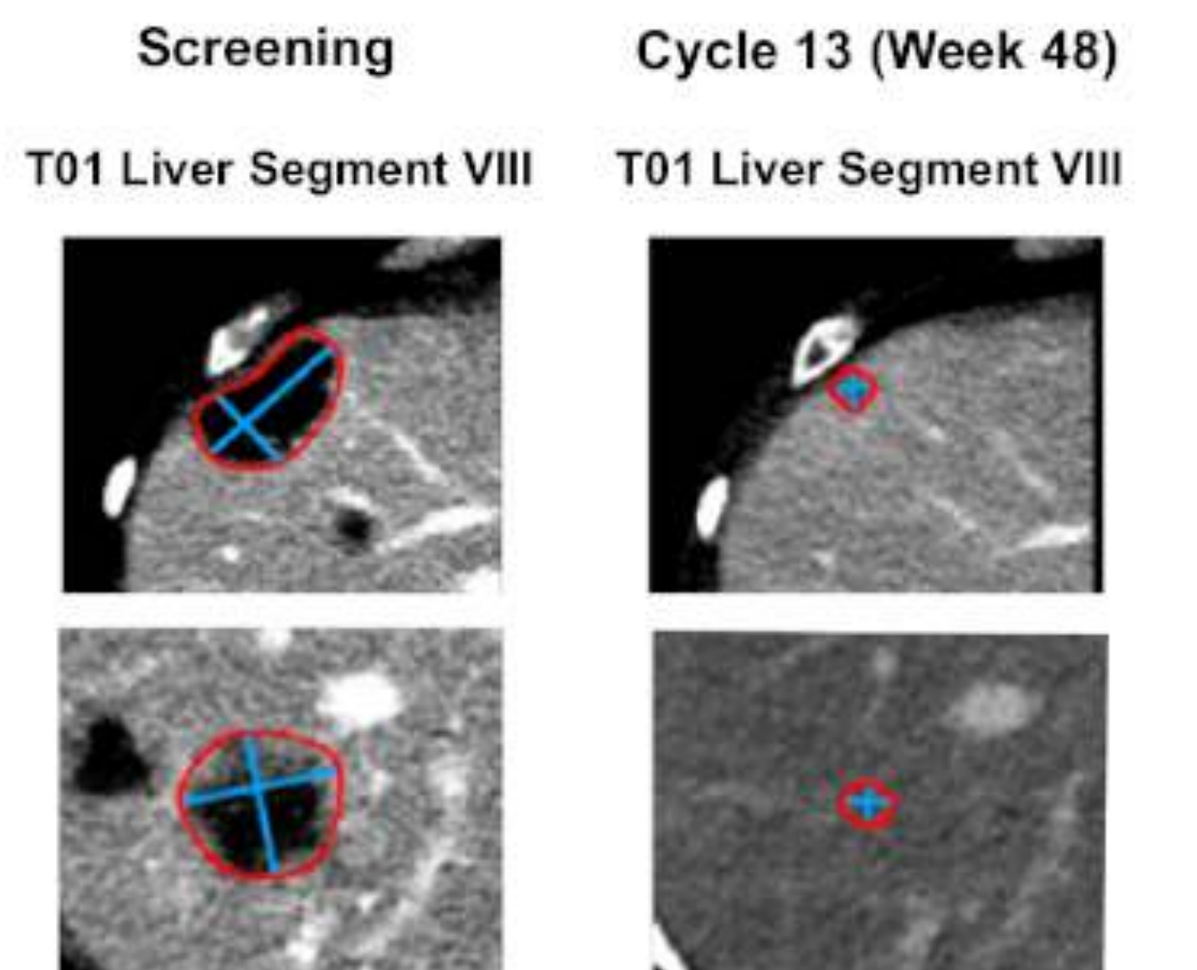
Screening
Cycle 13 (Week 48)
T01 Liver Segment VIII
T01 Liver Segment VIII

Patient # 09

- 51 year-old female
- Progression after 1st line chemotherapy
- PR by RECIST v 1.1 with a 47% reduction in SLD

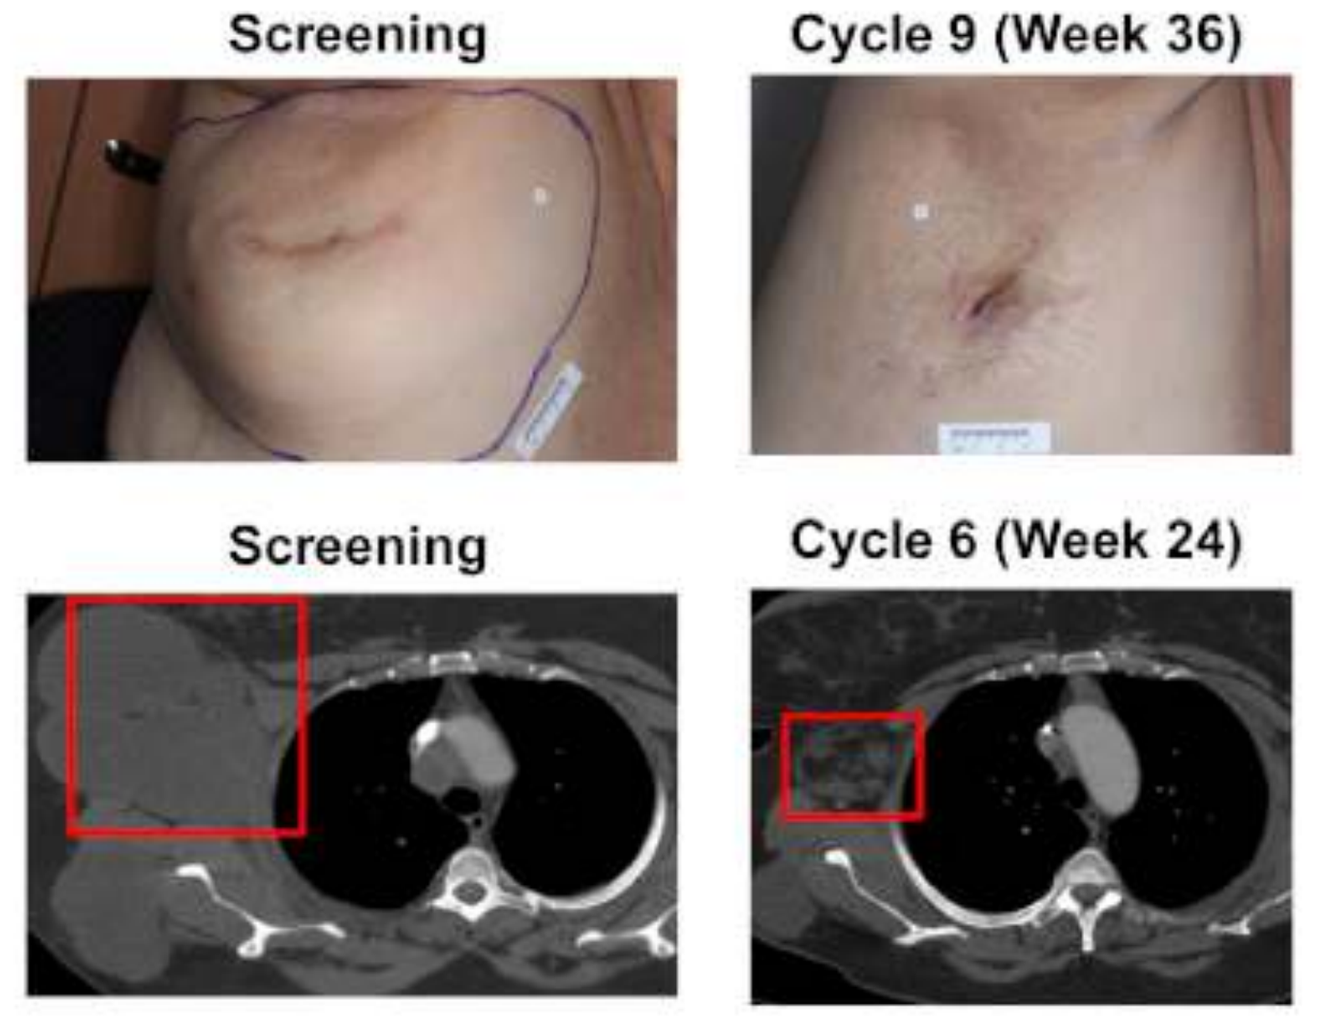
Screening
Cycle 9 (Week 36)
Screening
Cycle 6 (Week 24)

Patient # 13

- 46 year-old female
- Heavily pre-treated metastatic disease with 4 prior therapies for advanced disease
- PR by RECIST v1.1 with a 49% reduction in SLD
- Reduction in treated cutaneous disease and stable untreated lung metastasis

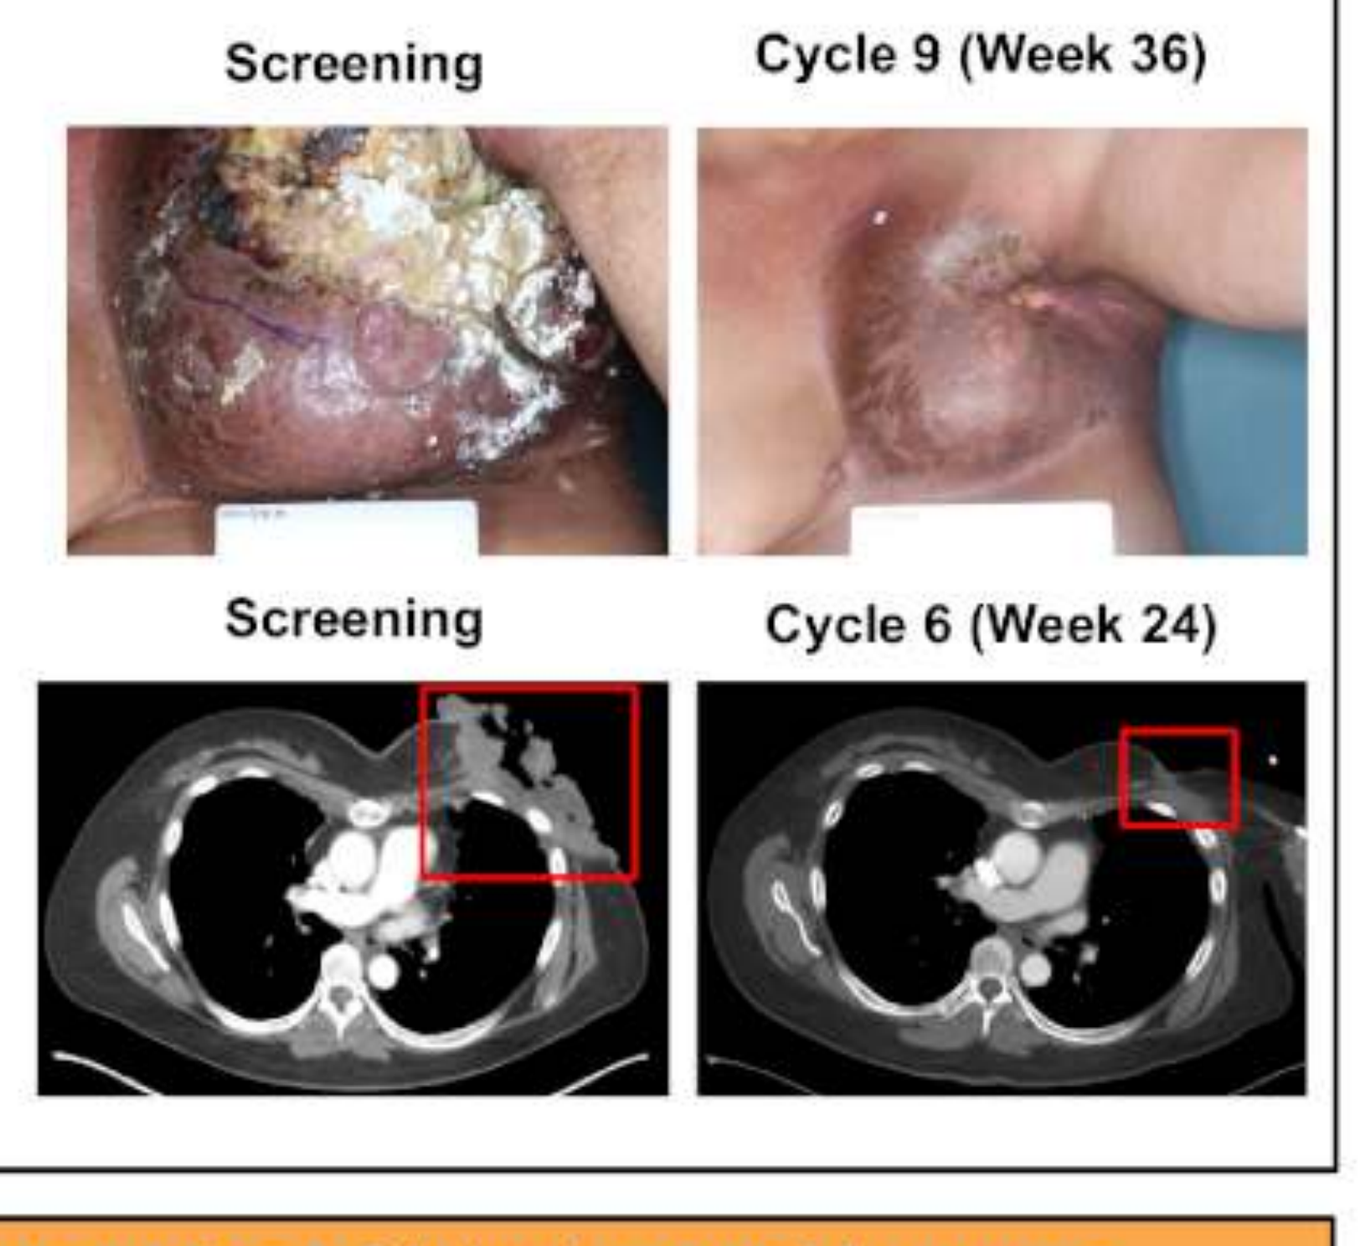
Screening
Cycle 9 (Week 36)
Screening
Cycle 6 (Week 24)

Figure 4: Low Frequencies of Intratumoral Immune Subsets at Screening (mIHC)

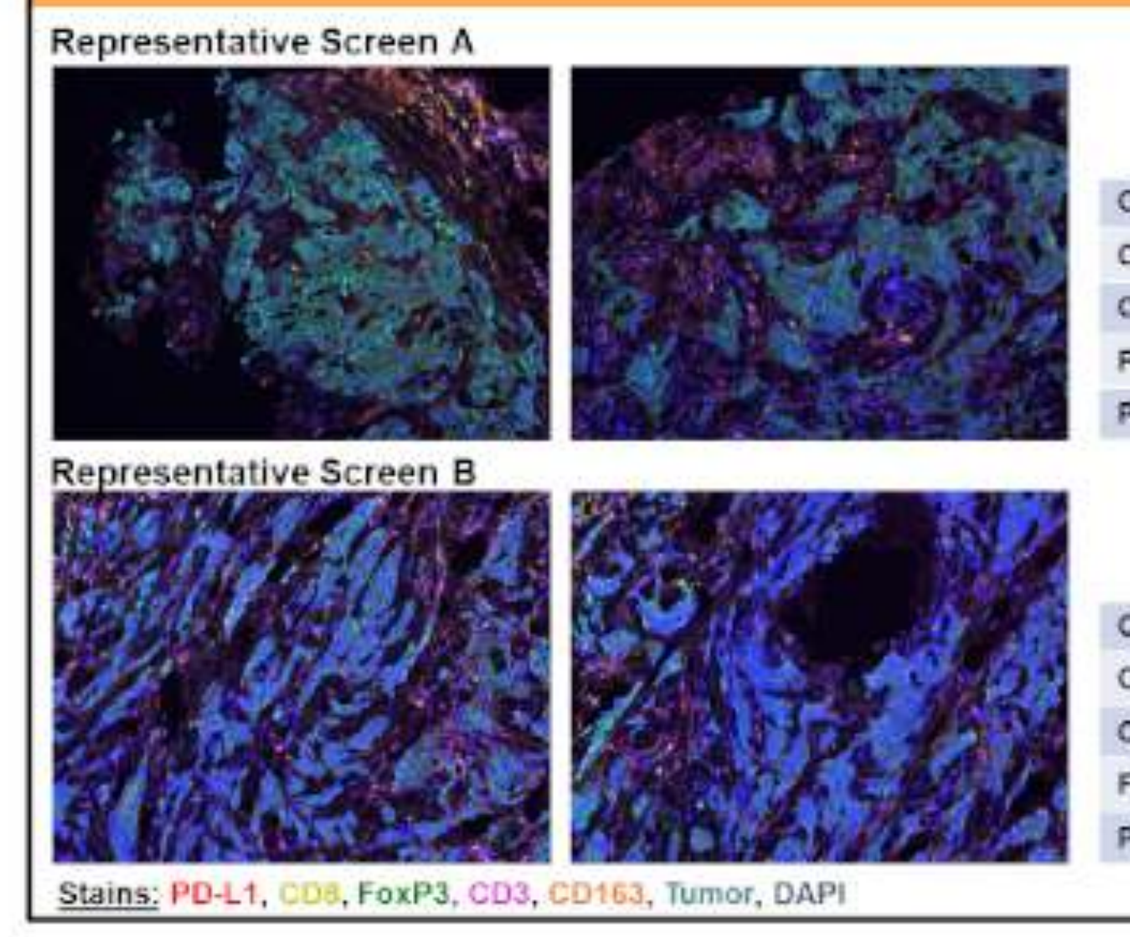
Representative Screen A
Representative Screen B
Stains: PD-L1, CD8, FoxP3, CD3, CD163, Tumor, DAPI

Representative Screen A

	Screen (cells/mm²)
CD8⁺	122.21
CD4⁺	90.45
CD163⁺	449.73
FoxP3⁺	6.15
PD-L1⁺	0.68

Representative Screen B

	Screen (cells/mm²)
CD8⁺	97.37
CD4⁺	119.05
CD163⁺	743.10
FoxP3⁺	20.16
PD-L1⁺	54.79

Figure 4: Multispectral IHC visualization of intratumoral immune subsets in representative biopsies taken at screening.

Enrolled patients have low frequencies of TIL (CD8⁺ and CD4⁺ T cells) with high relative amounts of suppressive CD163⁺ M2 macrophages.

Abbreviations: CNS, central nervous system; ECOG PS, Eastern Cooperative Oncology Group Performance Status; EP, electroporation; HER2, human epidermal growth factor receptor 2; HIV, human immunodeficiency virus; IHC, immunohistochemistry; PD-1, programmed cell death protein 1; RECIST, Response Evaluation Criteria in Solid Tumors; TNBC, triple negative breast cancer; P, pembrolizumab., ITT=intent to treat

Figure 5: Productive Immunological Changes in the Tumor Microenvironment and Periphery

Immune Response: Subjects demonstrated on-treatment peripheral and intratumoral immunological responses including significant increases in proliferating T cell subsets, decreases in markers associated with CPI non-response, increased levels of systemic chemokines, and increased IFN-γ-responsive transcriptome.

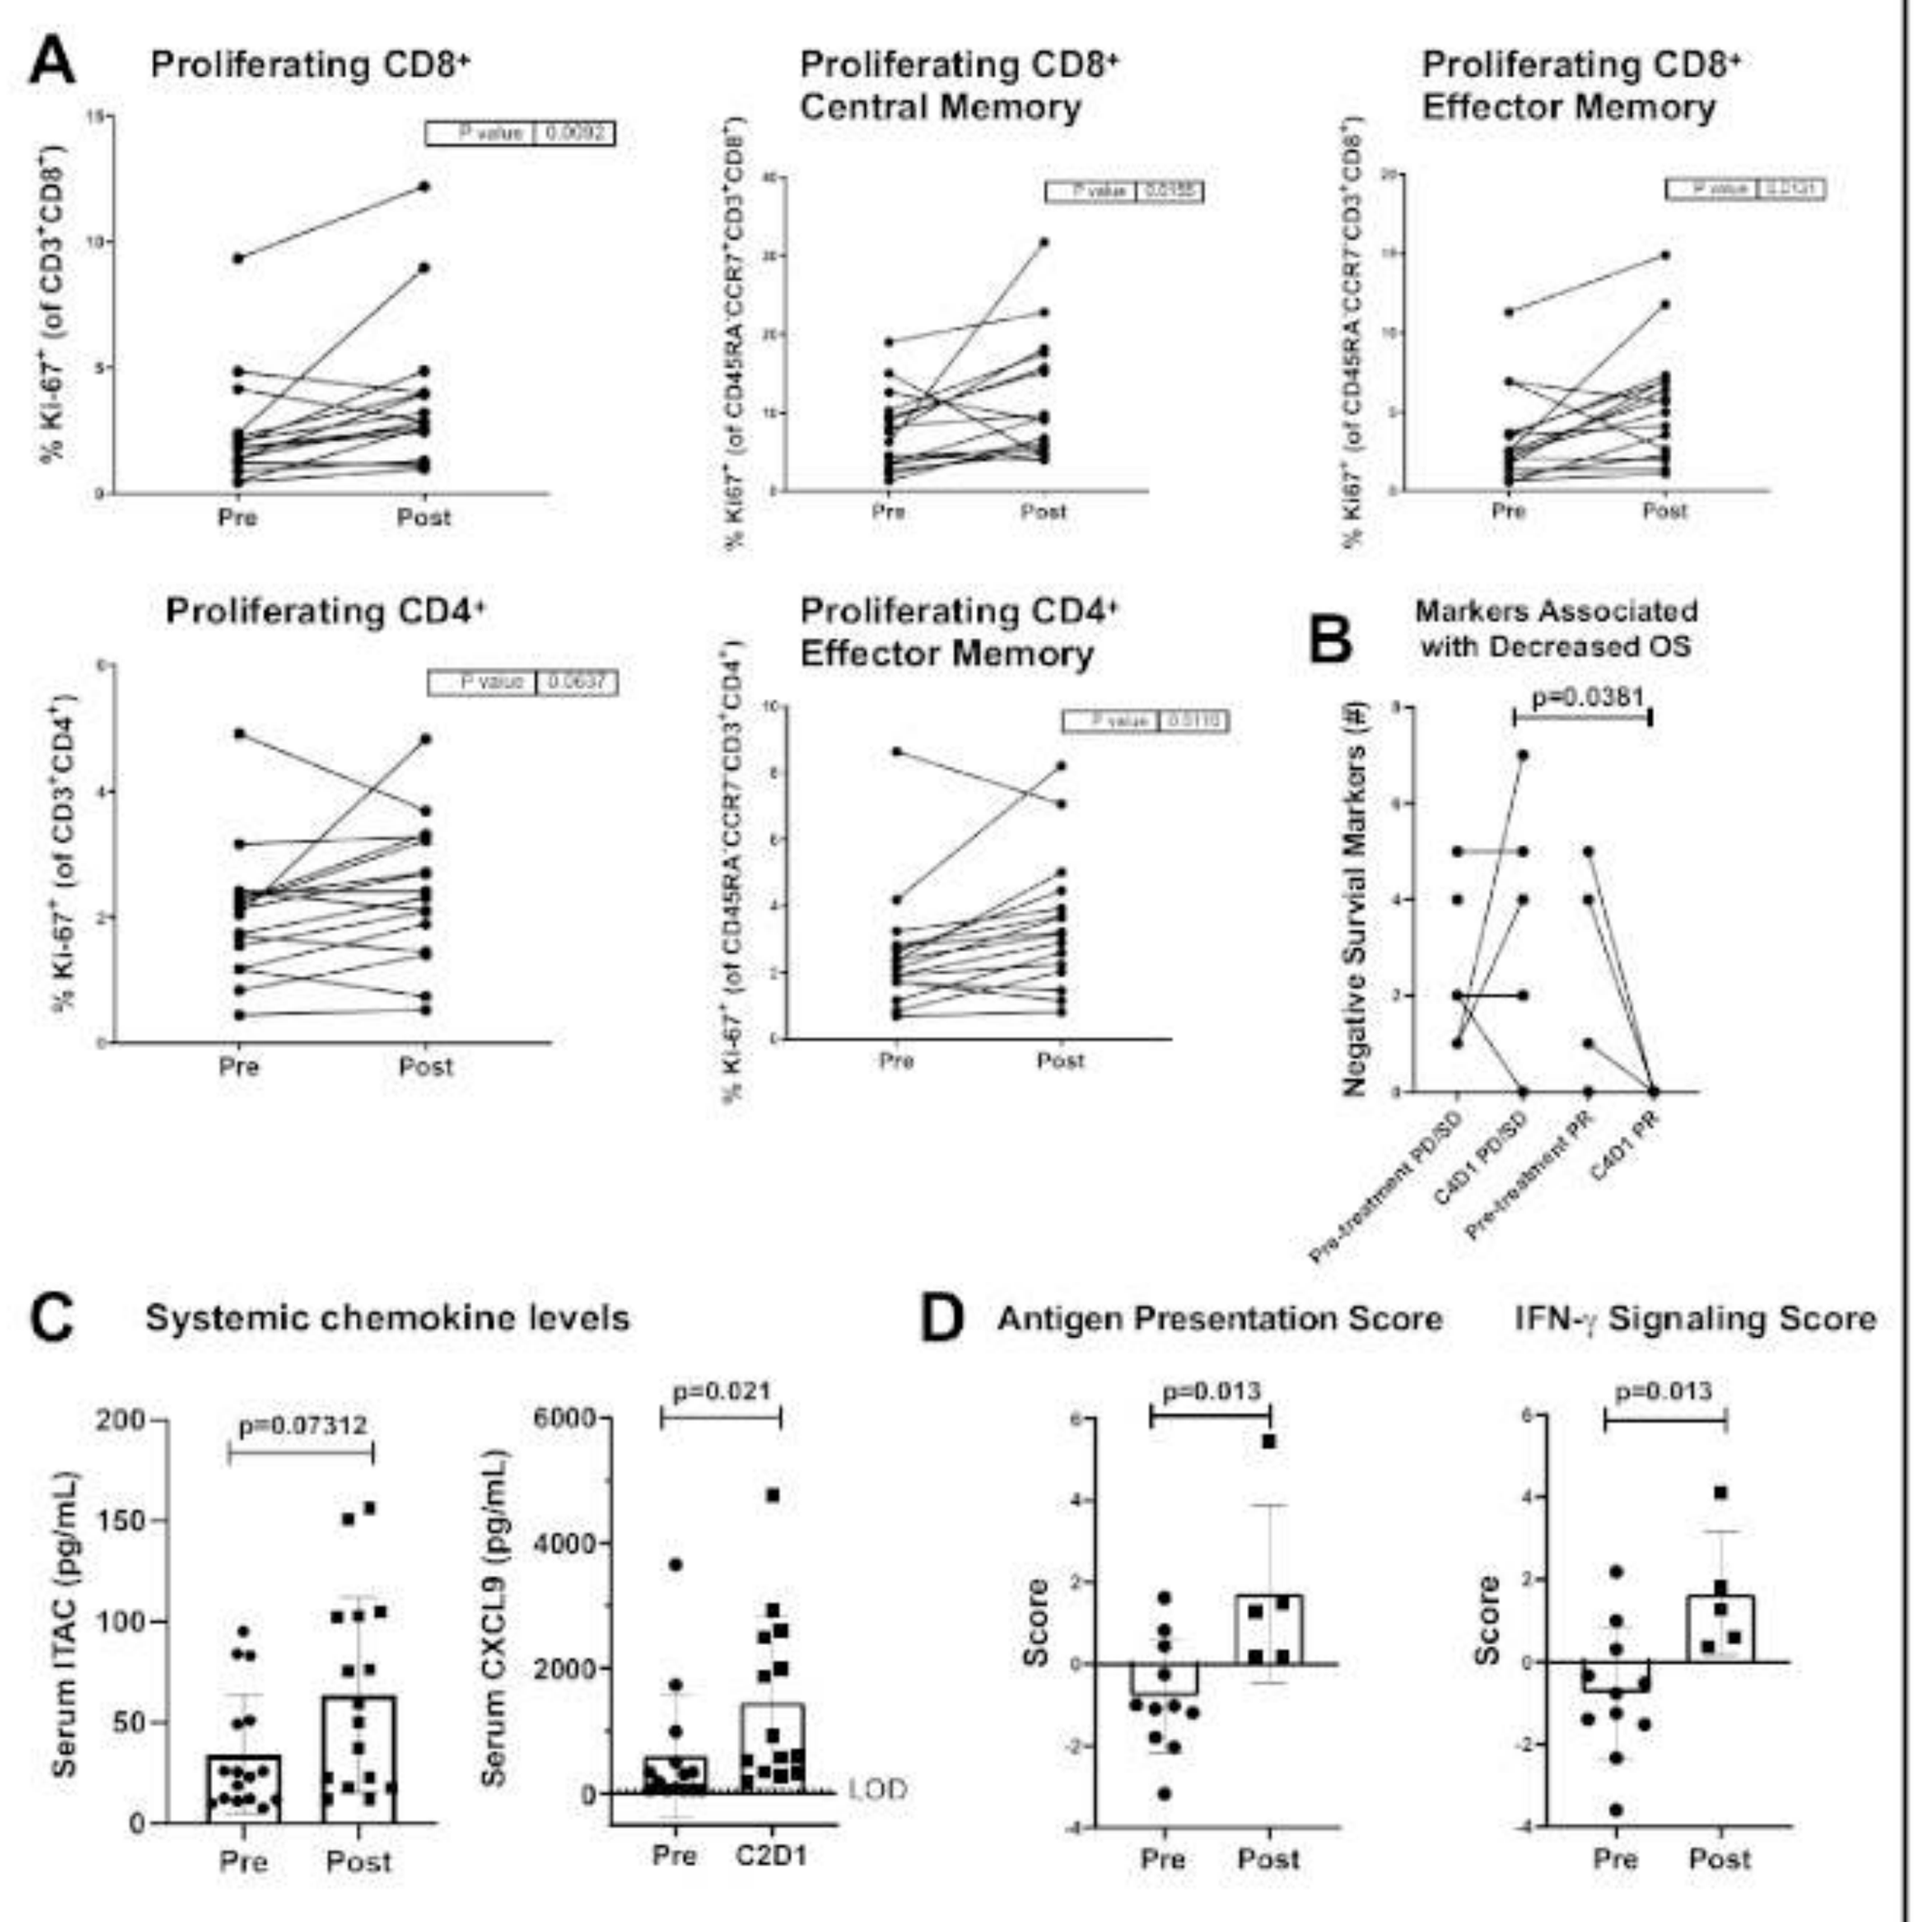
A
Proliferating CD8⁺
Proliferating CD8⁺ Central Memory
Proliferating CD8⁺ Effector Memory
Proliferating CD4⁺
Proliferating CD4⁺ Effector Memory
Pre
Post
B
Markers Associated with Decreased OS
p=0.0381
Negative Survival Markers (#)
Pre-treatment PD/SD
C4D1 PD/SD
Pre-treatment PR
C4D1 PR
C
Systemic chemokine levels
p=0.07312
Serum ITAC (pg/mL)
p=0.021
Serum CXCL9 (pg/mL)
LOD
Pre
C2D1
D
Antigen Presentation Score
IFN-γ Signaling Score
p=0.013
p=0.013
Score

Figure 5: **(A)** Peripheral immune subsets at screening (Pre) and post-treatment for all evaluable subjects. Significant increases in proliferating CD8 and CD4 subsets were observed. **(B)** Changes in blood safety markers[11,12,13,14] associated with poor response to CPI at Screen and on Cycle 4 for responding and non-responding subjects. Responding patients exhibit treatment-related decreases in negative survival markers. **(C)** Serum cytokine analysis showing post-treatment increases in ITAC and significant increases in CXCL9 (Luminex MAGPIX; R&D Systems DuoSet ELISA). **(D)** Treatment-related upregulation of immune-based transcripts in the tumor microenvironment related to antigen presentation and IFN-γ signaling (unmatched biopsies; NanoString IO360). Significant post-treatment changes were observed.

Summary and Conclusions

This interim analysis from KEYNOTE-890 suggests TAVO™ and pembrolizumab in combination has anti-tumor activity in heavily pre-treated metastatic TNBC patients.

- Intent-to-treat analysis yielded a 28.6% ORR (n=14)
 - 3 of the 4 responding subjects had PD-L1 status by IHC available; all 3 had tumors lacking PD-L1.
 - 3 of 4 responding subjects experienced deepening responses over 6 months
- TAVO™ and pembrolizumab are well tolerated, with only 3 of 16 subjects experiencing grade 3 treatment-related adverse events with combination treatment.
- Subjects demonstrated on-treatment immunological responses in the periphery, with significant increases in key proliferating T cell subsets, decreased markers associated with CPI non-response and increased systemic chemokines as well as increased intratumoral gene expression typically associated with productive responses such as increased antigen presentation and IFN-γ signatures.

P3-09-04

oncosec IMMUNOTHERAPIES

San Antonio Breast Cancer Symposium 2019

Contacts: OncoSec Kellie Malloy Foerter kmalloy@oncosec.com; Lead Investigator Melinda Telli, mtelli@stanford.edu